Phase 1 Results of CYT-0851 in Patients with Advanced Solid and Hematologic Cancers

Ryan C. Lynch[1], Pamela N. Munster[2], Ranjana Advani[3], Mehdi Hamadani[4], David R. Spigel[5], Gerald S Falchook[6], Manish R. Patel[7], David S. Siegel[8], Nina Beri[9], Grzegorz S. Nowakowski[10], Neil Palmisiano[11], Monika L. Burness[12], Kathleen N. Moore[13], Geoffrey Shapiro[14], Dejan Juric[15], William D. Bradley[16], Thomas J. O'Shea[16], Markus F. Renschler[16], Judson M. Englert[16], Timothy A. Yap[17]

[1]Fred Hutchinson Cancer Research Center, Seattle, WA; [2]University of California, San Francisco, San Francisco, CA; [3]Stanford Cancer Center, Stanford, CA; [4]Division of Hematology & Oncology, Medical College of Wisconsin, Milwaukee, WI; [5]Sarah Cannon Research Institute/Tennessee Oncology, Nashville, TN; [6] Sarah Cannon Research Institute at HealthONE, Denver, CO; [7]Sarah Cannon Research Institute/Florida Cancer Specialists, Sarasota, FL; [8]John Theurer Cancer Center, Hackensack University Medical Center, Hackensack, NJ; [9] Laura and Isaac Perlmutter Cancer Center, NYU Langone Health, New York, NY;[10]Division of Hematology, Mayo Clinic, Rochester, MN; [11]Sidney Kimmel Cancer Center, Thomas Jefferson University Hospital, Philadelphia, PA; [12]University of Michigan Rogel Cancer Center, Ann Arbor, MI; [13]University of Oklahoma Medical Center, Oklahoma City, OK; [14]Dana-Farber Cancer Institute, Boston, MA; [15]Massachusetts General Hospital, Boston, MA; [16]Cyteir Therapeutics, Lexington, MA; [17] The University of Texas MD Anderson Cancer Center, Houston, TX

Abstract # 3084

Background and Rationale

CYT-0851 was discovered using a phenotypic screen designed to exploit a novel synthetic lethality between constitutive overexpression of the cytidine deaminase enzyme, activation-induced cytidine deaminase (AID), and impairment of the double strand deoxyribonucleic acid (DNA) repair pathway, homologous recombination (HR). As a result of molecular, bioinformatic, and biochemical characterization, we now believe that the anti-cancer effects of CYT-0851 are primarily the result of inhibition of monocarboxylate transporter (MCT) activity and impairment of lactate transport.

Figure 1: MCTs are Membrane Transporters Fundamental to Cancer Survival

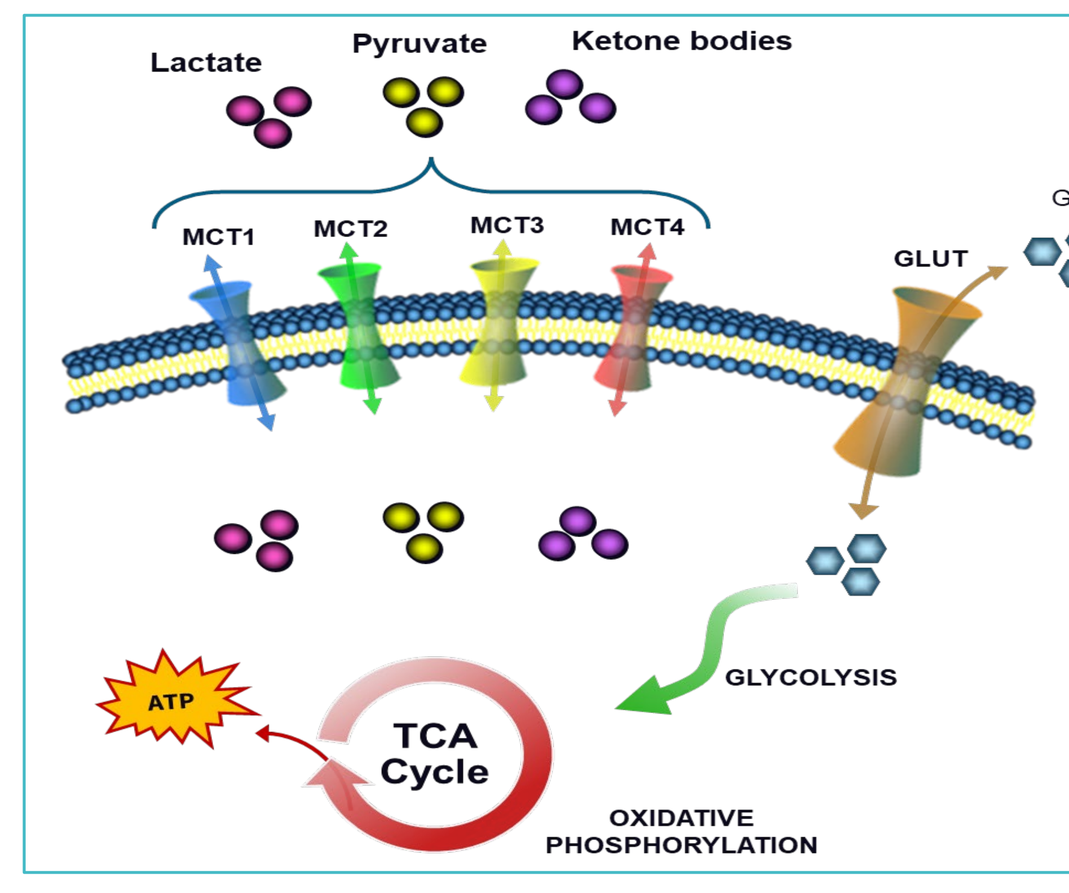
Lactate
Pyruvate
Ketone bodies
MCT1
MCT2
MCT3
MCT4
GLUT
Glucose
GLYCOLYSIS
ATP
TCA Cycle
OXIDATIVE PHOSPHORYLATION

MCTs are plasma membrane proteins that bi-directionally transport monocarboxylated biomolecules such as lactate, pyruvate, and ketone bodies and are often upregulated in cancers where they predict poor patient prognosis and increased patient mortality **(Figure 1).** Metabolic reprogramming is a hallmark of cancer and is characterized by increased dependence on lactate-producing glycolysis to support the metabolic needs of ongoing tumorigenesis. Inhibiting MCT function in glycolytic cancer cells leads to an accumulation of intracellular lactate that impairs glycolysis and inhibits tumor cell growth **(Figure 2)**, thereby making MCTs an attractive target for cancer therapy.

Figure 2: CYT-0851 Induced Intracellular Lactate Accumulation Correlates with Cytotoxicity in Both Hematologic and Solid Cancer Cell Lines

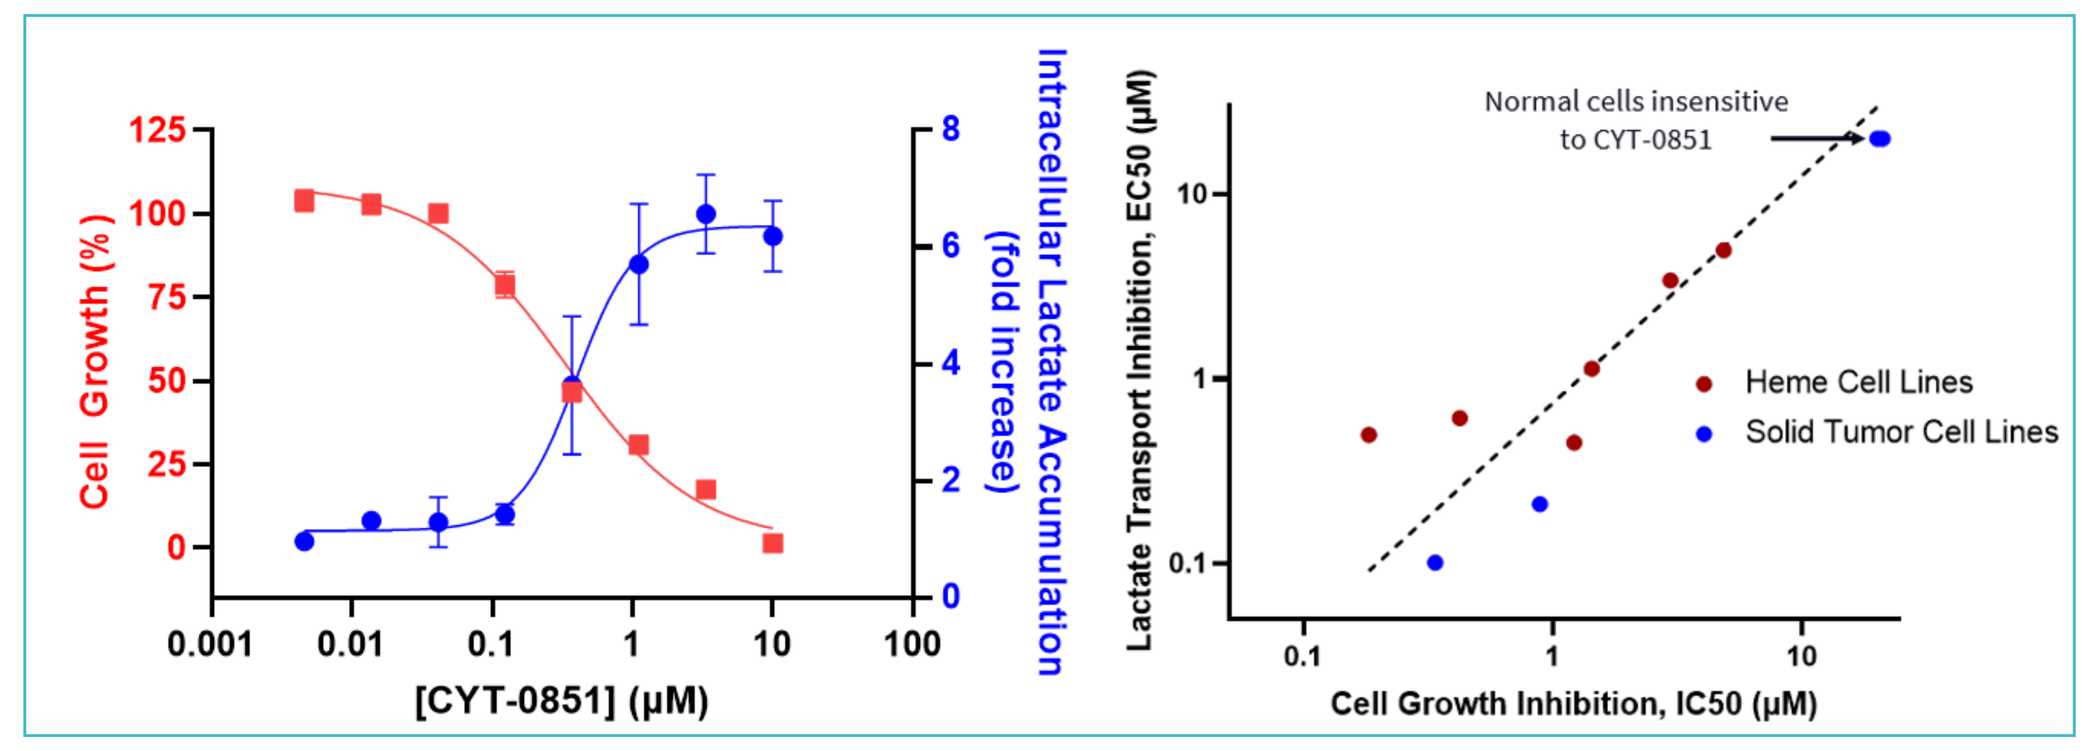
Cell Growth (%)
Intracellular Lactate Accumulation (fold increase)
[CYT-0851] (µM)
Lactate Transport Inhibition, EC50 (µM)
Cell Growth Inhibition, IC50 (µM)
Normal cells insensitive to CYT-0851
Heme Cell Lines
Solid Tumor Cell Lines

Inhibition of MCT-mediated lactate transport indirectly downregulates several cell-cycle regulated proteins, including the HR effector protein, RAD51, which we believe may explain the observed preclinical effects of CYT-0851 on DNA repair.

Study Objectives

Primary	To determine the recommended Phase 2 dose (RP2D) and maximum-tolerated dose (MTD)
Secondary	To evaluate the safety and tolerability
	To determine the pharmacokinetic parameters and optimal dosing regimen (BID versus QD)
	To characterize the preliminary anti-tumor activity

Methods – Study Design

Eligibility Criteria

Key Inclusion Criteria	Key Exclusion Criteria
• ECOG 0-1 • Measurable disease by disease-specific criteria • Relapsed/refractory B-cell malignancies • Non-Hodgkin Lymphoma, CLL, Multiple myeloma • Advanced solid tumors • Breast, HNSCC, ovarian, soft-tissue sarcoma, Small- cell lung cancer (SCLC) and pancreatic cancer	• Prior allogeneic stem cell transplant • Absolute neutrophil count < 1.0 × 10^9/L • Platelets < 75 × 10^9/L • Hemoglobin < 9.0 g/dL • Creatinine Clearance < 40 mL/min • AST/ALT > 2.0 x ULN

Phase 1 Study Design

CYT-0851 was administered orally once or twice daily on a 28-day schedule (1 Cycle)

Dose-escalation proceeded via accelerated titration then 3+3 design; Starting Dose = 15 mg BID
Up to 12 patients were backfilled into each cleared dose level

Treatment was administered until disease progression, unacceptable toxicity, or withdrawal

Response assessments were conducted every 2 cycles (8 weeks) by disease-specific criteria (RECIST v1.1 (solid tumor) and Lugano (NHL))

Intra-patient dose-escalation was allowed to the highest cleared dose after 1 cycle
Patients are analyzed by the dose level in which they enrolled.

Results

Demographics and Disposition

- Enrollment started on September 30, 2019 and is ongoing at 18 US sites
- As of a data cutoff of April 13, 2022, 80 patients have been enrolled in Ph1 across 12 dose-levels

Table 1: Demographics and Characteristics

Characteristics	Total (n=80) n (%)
Female	38 (47.5)
Male	42 (52.5)
Median age (range)	63 (34-87)
ECOG Performance Status	
0	22 (27.5)
1	56 (70.0)
Prior Lines of Therapy	
Median (range)	4 (1-13)
1	2 (2.5)
2	15 (18.8)
≥ 3	63 (78.8)
Tumor Type	
Breast Cancer	8 (10.0)
Head and Neck Cancer	7 (8.8)
SCLC	4 (5.0)
Mucoepidermoid Cancer	1 (1.3)
Multiple Myeloma	3 (3.8)
NHL	21 (26.3)
Ovarian Cancer	4 (5.0)
Pancreatic Cancer	14 (17.5)
Soft-Tissue Sarcoma	18 (22.5)

Figure 3: Patient Enrollment and Disposition

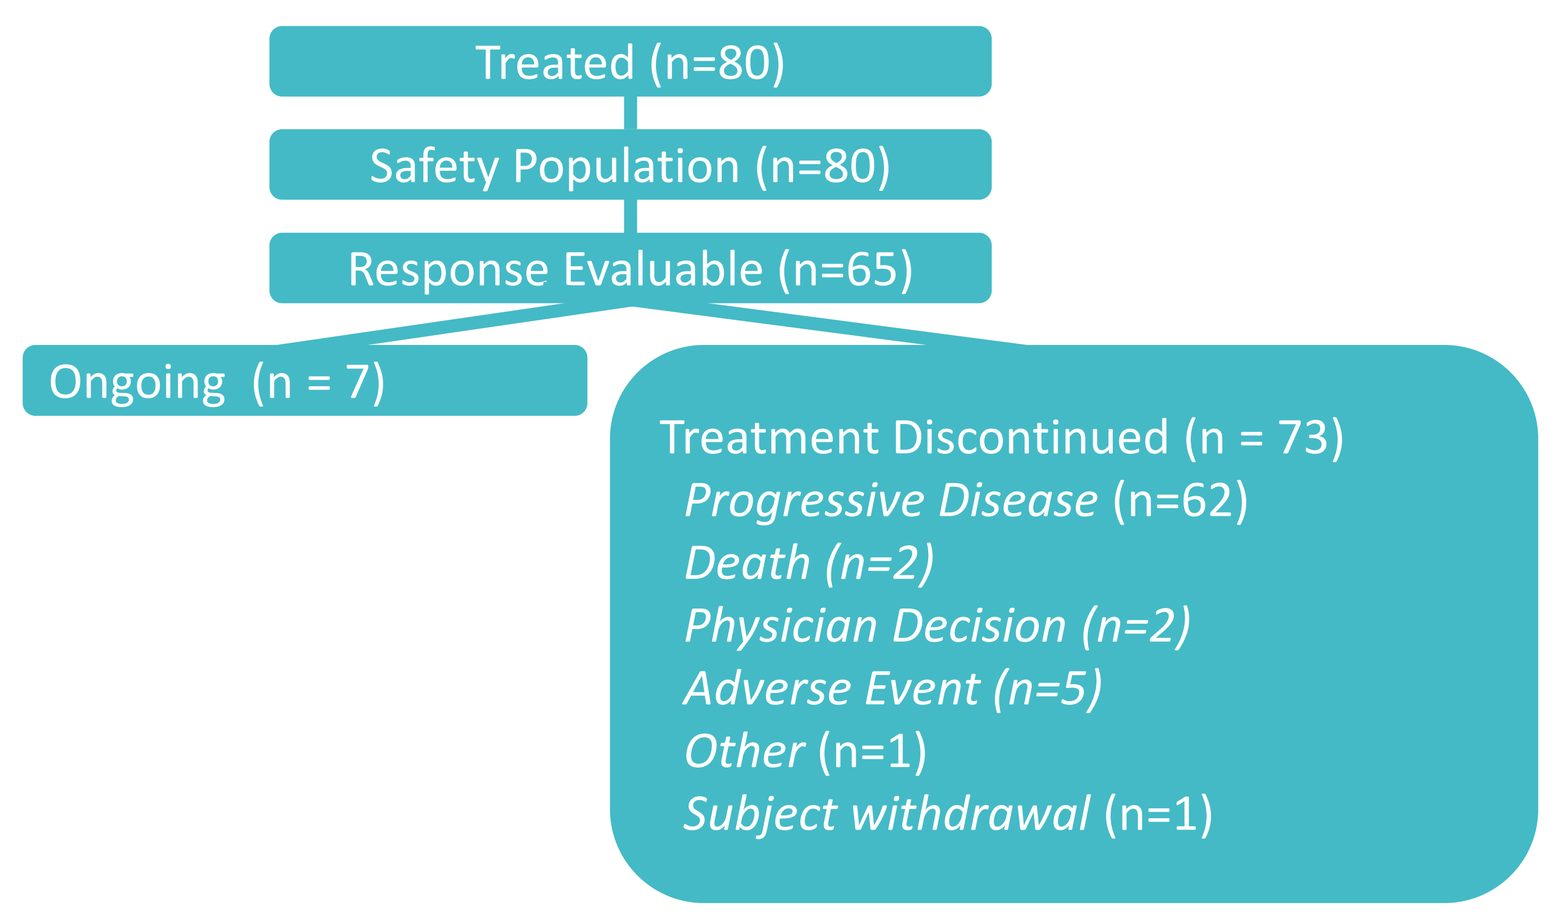
Treated (n=80)
Safety Population (n=80)
Response Evaluable (n=65)
Ongoing (n = 7)
Treatment Discontinued (n = 73)
Progressive Disease (n=62)
Death (n=2)
Physician Decision (n=2)
Adverse Event (n=5)
Other (n=1)
Subject withdrawal (n=1)

Table 2: Enrollment by Dose Level

	CYT-0851 Dose Level												
	15 mg BID	20 mg BID	30 mg BID	45 mg BID	90 mg QD	130 mg QD	200 mg QD	300 mg QD	400 mg QD	600 mg QD	400 mg BID	600 mg BID	Total
N	1	1	4	7	6	11	10	12	10	11	4	3*	80

*1 patient enrolled in the 600 mg BID cohort only received treatment with 400 mg QD due to the evolving safety profile and protocol changes.

Dose-Escalation and Determination of MTD

- Dose-limiting toxicities (DLTs) and adverse events impacting tolerability and dosing were monitored in all pts
- 600 mg BID: 1 of 2 treated pts (50%) experienced a DLT
- 400 mg BID: 2 of 4 pts (50%) experienced TRAEs in cycle 2 requiring modification and it was declared intolerable
- 600 mg QD: 3 of 11 pts (27%) experienced TRAEs requiring dose modification, and this was declared the MTD

Table 3: DLTs and Treatment-Related Adverse Events Requiring Dose Modification

Dose Level	n	n with DLTs	TRAE requiring modification	Description of Adverse Event
600 mg BID	2*	1	0	Gr 3 Acidosis (Starvation Ketoacidosis)
400 mg BID	4	0	2	Dose reduced: Grade 3 Polyarthropathy, Body Rash and Myalgia Dose withdrawn: Gr 3 Dry skin
600 mg QD	11	0	3	Dose reduced: Gr 3 Fatigue (2 patients) Dose withdrawn: Gr 3 Mucositis, Vomiting, Anorexia (1 Patient)

Safety

- TRAEs: 58% of pts reported any grade events and only 18% have reported Gr3/4 across all cohorts
- The most common TRAEs were fatigue (18%), alopecia (14%), nausea (11%), hyperuricemia (10%), constipation (8%) and anemia (8%)
- No treatment-related deaths have been reported

Table 4: Safety Overview by Dose Level

	15 mg BID (N=1)	20 mg BID (N=1)	30 mg BID (N=4)	45 mg BID (N=7)	90 mg QD (N=6)	130 mg QD (N=11)	200 mg QD (N=10)	300 mg QD (N=12)	400 mg QD (N=10)	600 mg QD (N=11)	400 mg BID (N=4)	600 mg BID (N=3)	Total (N=80)
Subjects With at Least One Related TEAE	1 (100.0%)	0 (0.0%)	2 (50.0%)	4 (57.1%)	4 (66.7%)	3 (27.3%)	4 (40.0%)	6 (50.0%)	8 (80.0%)	8 (72.7%)	4 (100.0%)	2 (66.7%)	46 (57.5%)
Subjects With at Least One Related Grade>=3 TEAE	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	1 (16.7%)	1 (9.1%)	1 (10.0%)	1 (8.3%)	3 (30.0%)	3 (27.3%)	3 (75.0%)	1 (33.3%)	14 (17.5%)
Subjects With at Least One Related Serious TEAE	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	1 (10.0%)	1 (9.1%)	1 (25.0%)	1 (33.3%)	4 (5.0%)
Subjects With at Least One Related TEAE Leading to Drug Withdrawal	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	1 (9.1%)	1 (25.0%)	0 (0.0%)	2 (2.5%)
Subjects With at Least One Related TEAE Leading to Drug Interruption	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	2 (33.3%)	0 (0.0%)	0 (0.0%)	1 (8.3%)	1 (10.0%)	0 (0.0%)	2 (50.0%)	2 (66.7%)	8 (10.0%)
Subjects With at Least One Related TEAE Leading to Dose Reduction	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	1 (16.7%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	2 (18.2%)	1 (25.0%)	0 (0.0%)	4 (5.0%)
Subjects With at Least One DLT	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	0 (0.0%)	1 (33.3%)	1 (1.3%)

Figure 4: Summary of Treatment-Emergent Adverse Events occurring in ≥ 10 % of pts by Relatedness

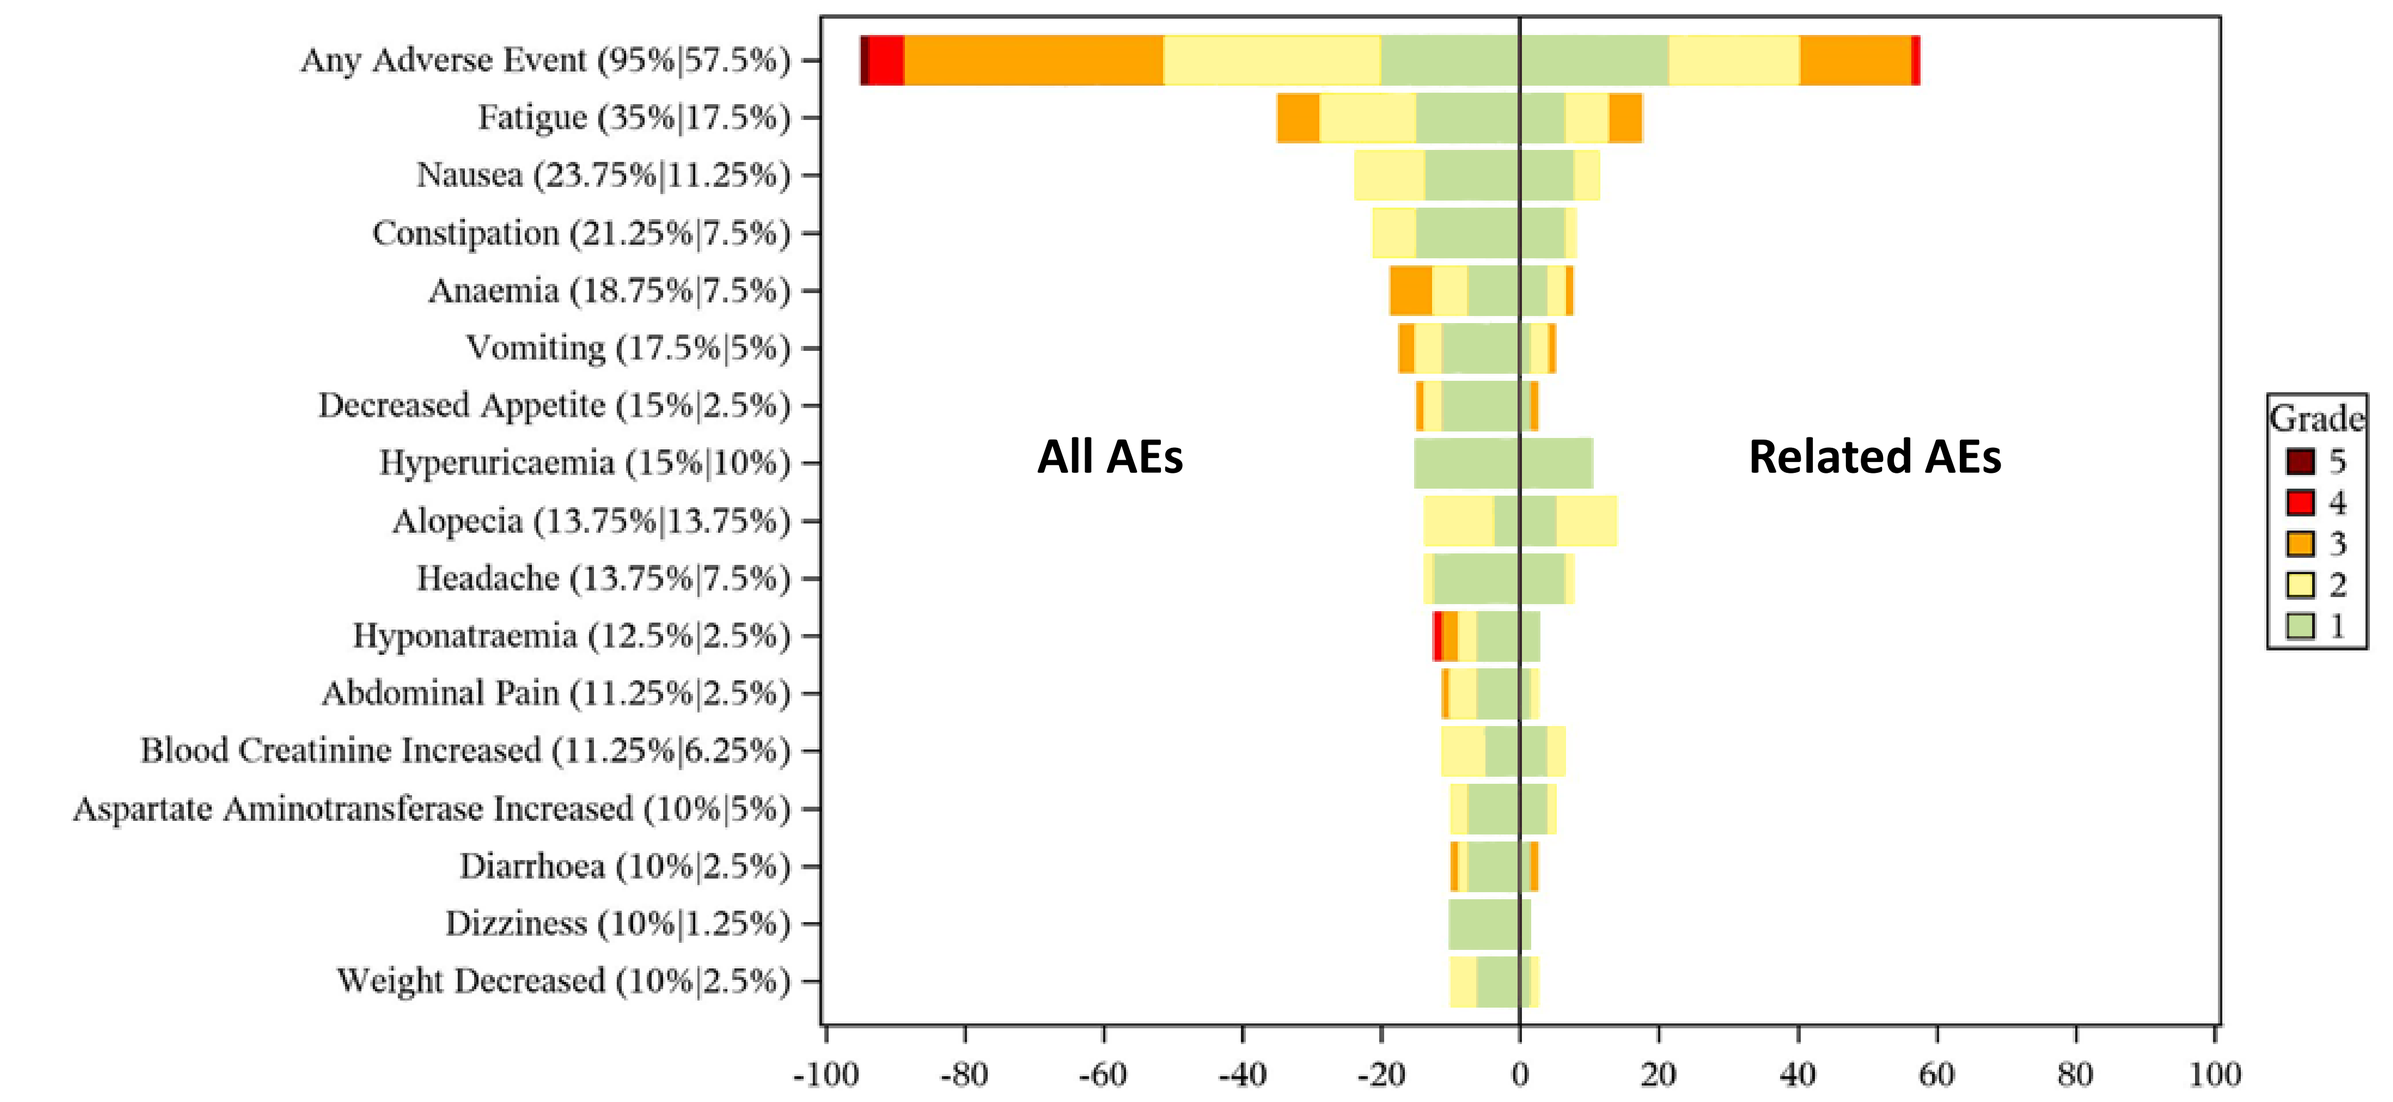
Any Adverse Event (95%|57.5%)
Fatigue (35%|17.5%)
Nausea (23.75%|11.25%)
Constipation (21.25%|7.5%)
Anaemia (18.75%|7.5%)
Vomiting (17.5%|5%)
Decreased Appetite (15%|2.5%)
Hyperuricaemia (15%|10%)
Alopecia (13.75%|13.75%)
Headache (13.75%|7.5%)
Hyponatraemia (12.5%|2.5%)
Abdominal Pain (11.25%|2.5%)
Blood Creatinine Increased (11.25%|6.25%)
Aspartate Aminotransferase Increased (10%|5%)
Diarrhoea (10%|2.5%)
Dizziness (10%|1.25%)
Weight Decreased (10%|2.5%)
All AEs
Related AEs
Grade
5
4
3
2
1

Pharmacokinetic Analysis

- Exposure of CYT-0851 exhibits approximately dose proportional behavior
- Effective half-life of ~3 days, derived from accumulation factor, supports QD administration
- Preliminary evaluation of food effect indicates no significant impact on exposure
- Mean steady-state trough levels achieved at RP2D exceed levels where preclinical studies demonstrated tumor growth inhibition

Figure 5: Pharmacokinetic Profiles Across All Dose Levels and 400 mg QD

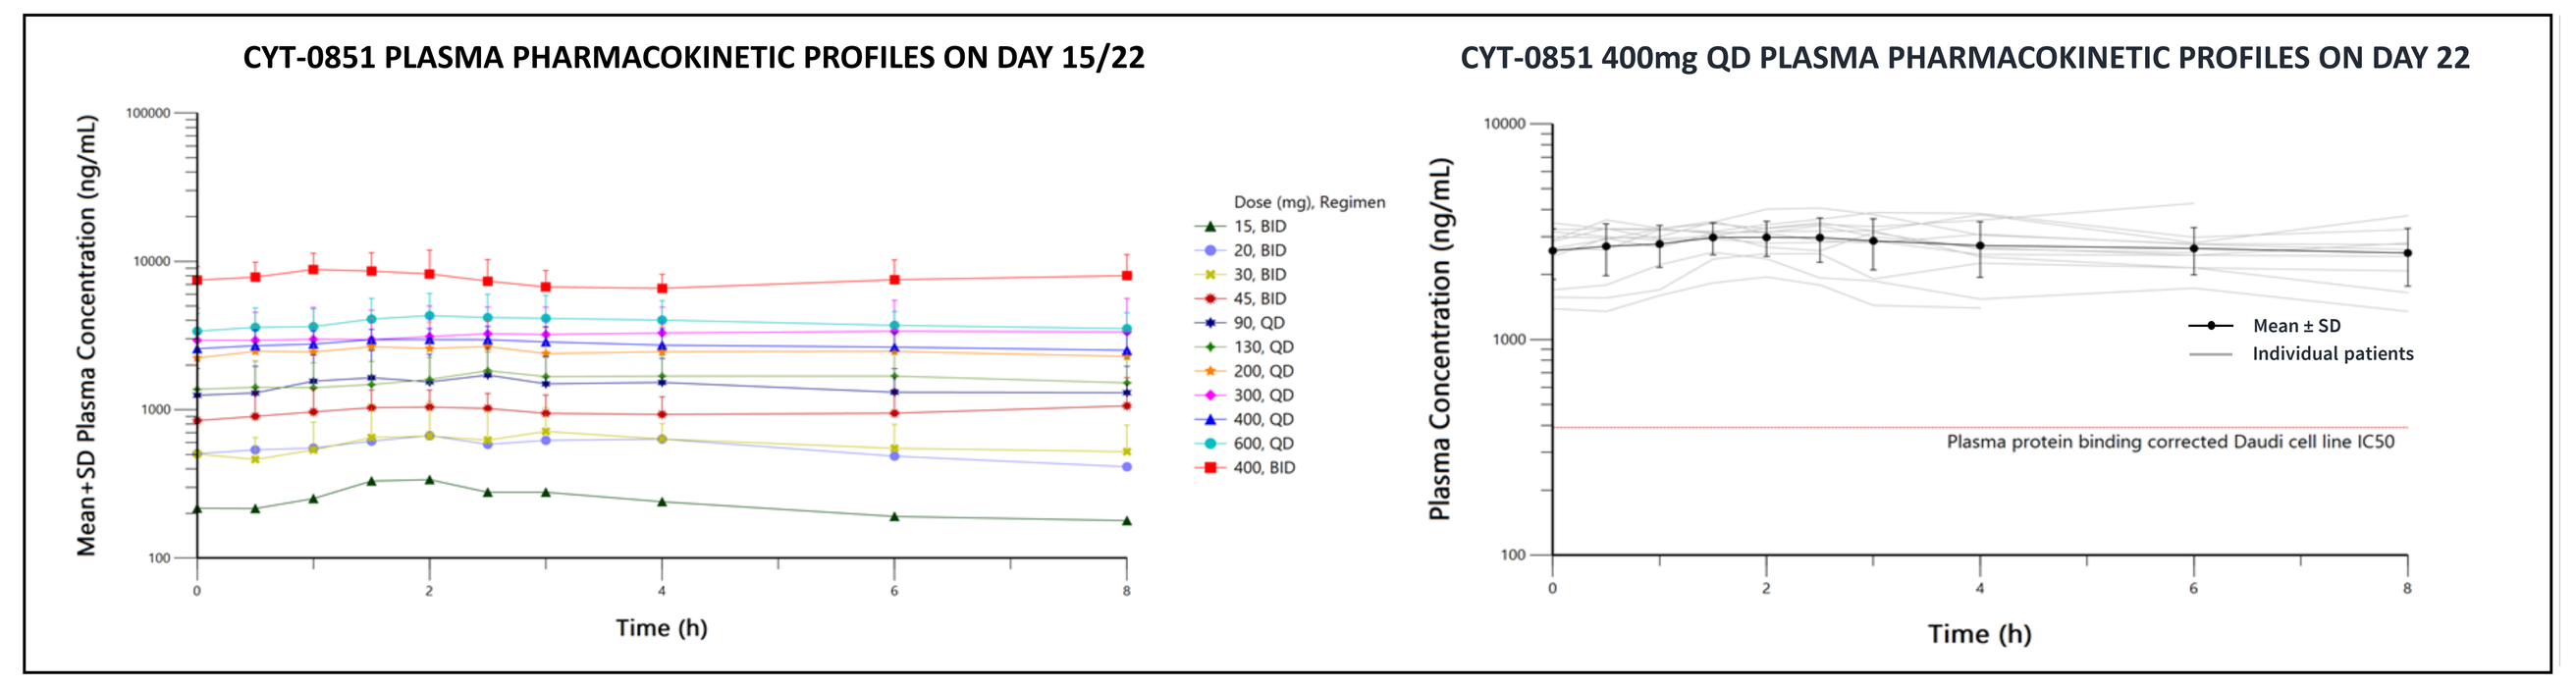
CYT-0851 PLASMA PHARMACOKINETIC PROFILES ON DAY 15/22
CYT-0851 400mg QD PLASMA PHARMACOKINETIC PROFILES ON DAY 22
Mean+SD Plasma Concentration (ng/mL)
Plasma Concentration (ng/mL)
Time (h)
Mean ± SD
Individual patients
Plasma protein binding corrected Daudi cell line IC50

Table 5: Summary of Pharmacokinetic Parameters Across Dose Levels

CYT-0851 Human Pharmacokinetics Summary for Day 15/22

Pharmacokinetic Parameter	Dose (mg) 15 BID (n=1)	20 BID (n=1)	30 BID (n=3)	45 BID (n=6)	90 QD (n=3)	130 QD (n=11)	200 QD (n= 9)	300 QD (n= 14)	400 QD (n=9)	600 QD (n=9)	400 BID (n=4)
T_{max} (h)	2.0	2.0	4 (2, 4)	2.5 (1.5, 8)[a]	2.5 (1, 2.5)[a]	2.5(1.5, 8)[a]	2.0 (1.5, 8)[a]	3.5 (2, 8)[a]	2.25 (0, 6)[a]	2.5(1.5, 6)[a]	1.75 (1, 8)[a]
C_{max} (ng/mL)	338	670	710 ± 390[b]	1200 ± 509[b]	1550 ± 930[b]	1830 ± 725[b]	2630± 1440[b]	3340 ± 2230[b]	3240 ± 696[b]	4460± 1650[b]	8950± 3180[b]
AUC_{0-8} (ng.h/mL)	1890	4410	4420 ± 2140[b]	7400 ± 2820[b]	10500 ± 5570[b]	12200± 5510[b]	17400 ± 9670[b]	22600± 14700[b]	21500± 4250[b]	29500± 9620[b]	58300± 19500[b]

[a]: median (min, max) [b]: geometric mean ± standard deviation (SD)

Clinical Activity in Solid Tumors

Figure 6: Waterfall Plot of Best Percentage Change in Tumor Size

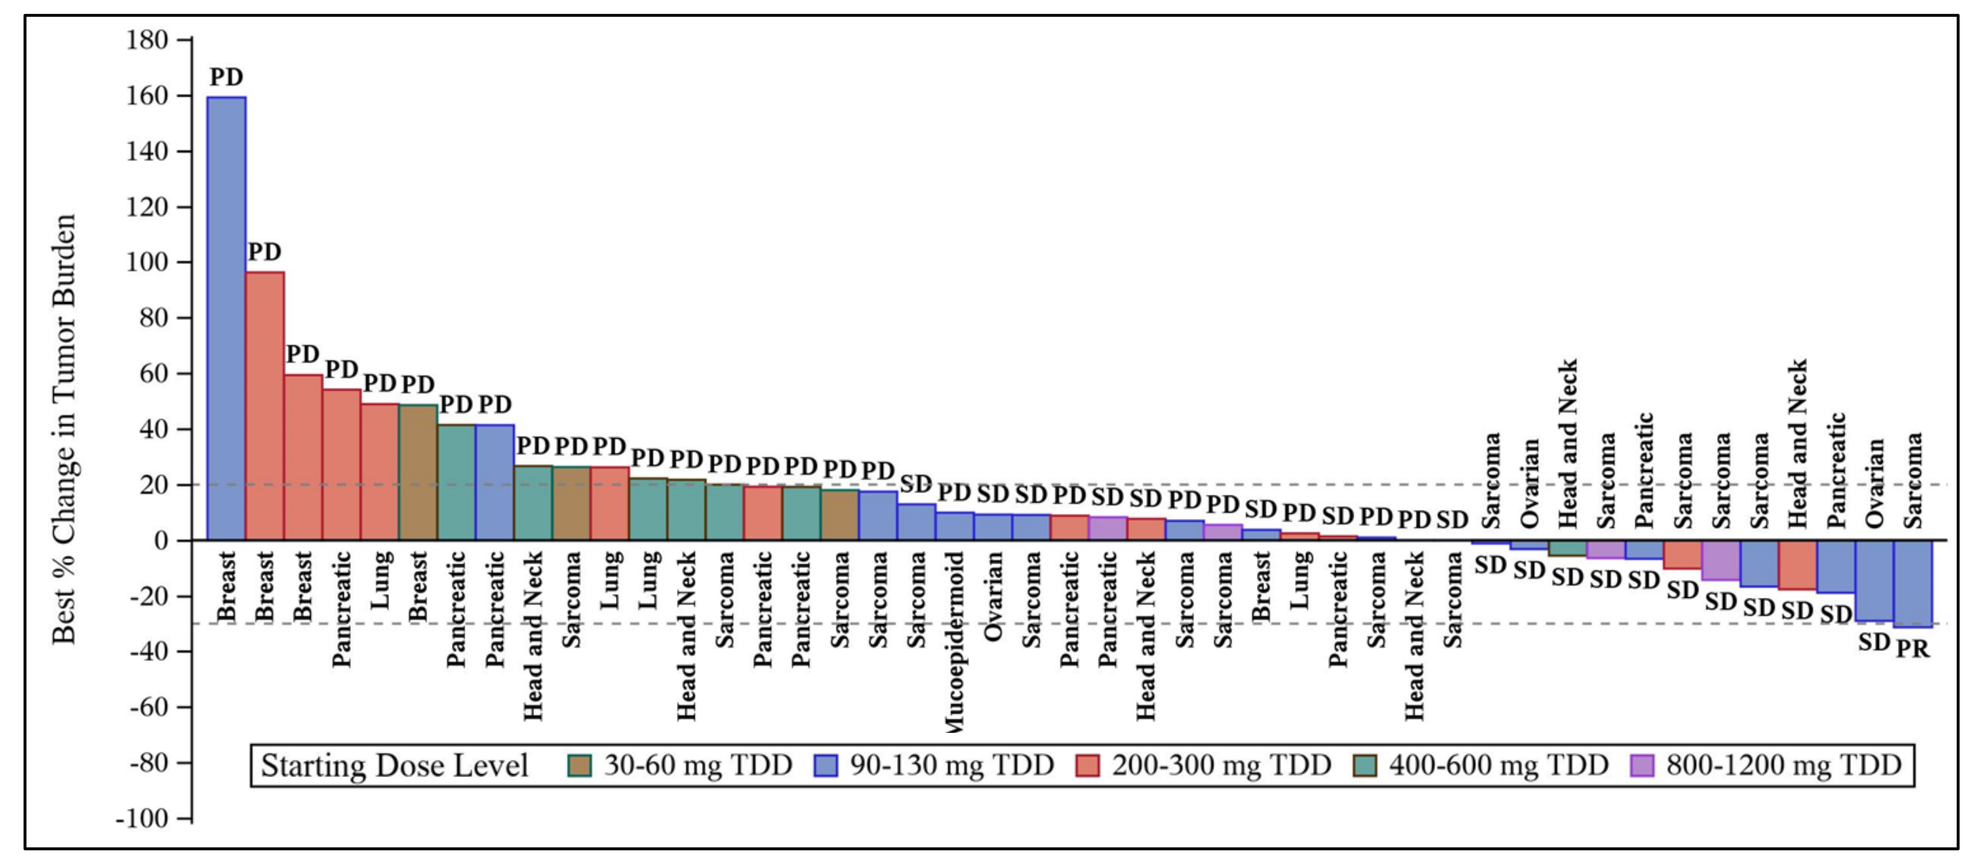
Best % Change in Tumor Burden
Starting Dose Level
30-60 mg TDD
90-130 mg TDD
200-300 mg TDD
400-600 mg TDD
800-1200 mg TDD

- 45 pts with solid tumors were response evaluable
- 1 unconfirmed PR in a pt with soft-tissue sarcoma treated for 11 cycles
- 19 pts (42%) had SD and 12 pts (27%) had a decrease in target lesion size
- The overall disease control rate was 44%
- 17 pts received treatment for ≥ 4 cycles

Figure 7: Swimmer's Plot of Duration of Treatment and Response

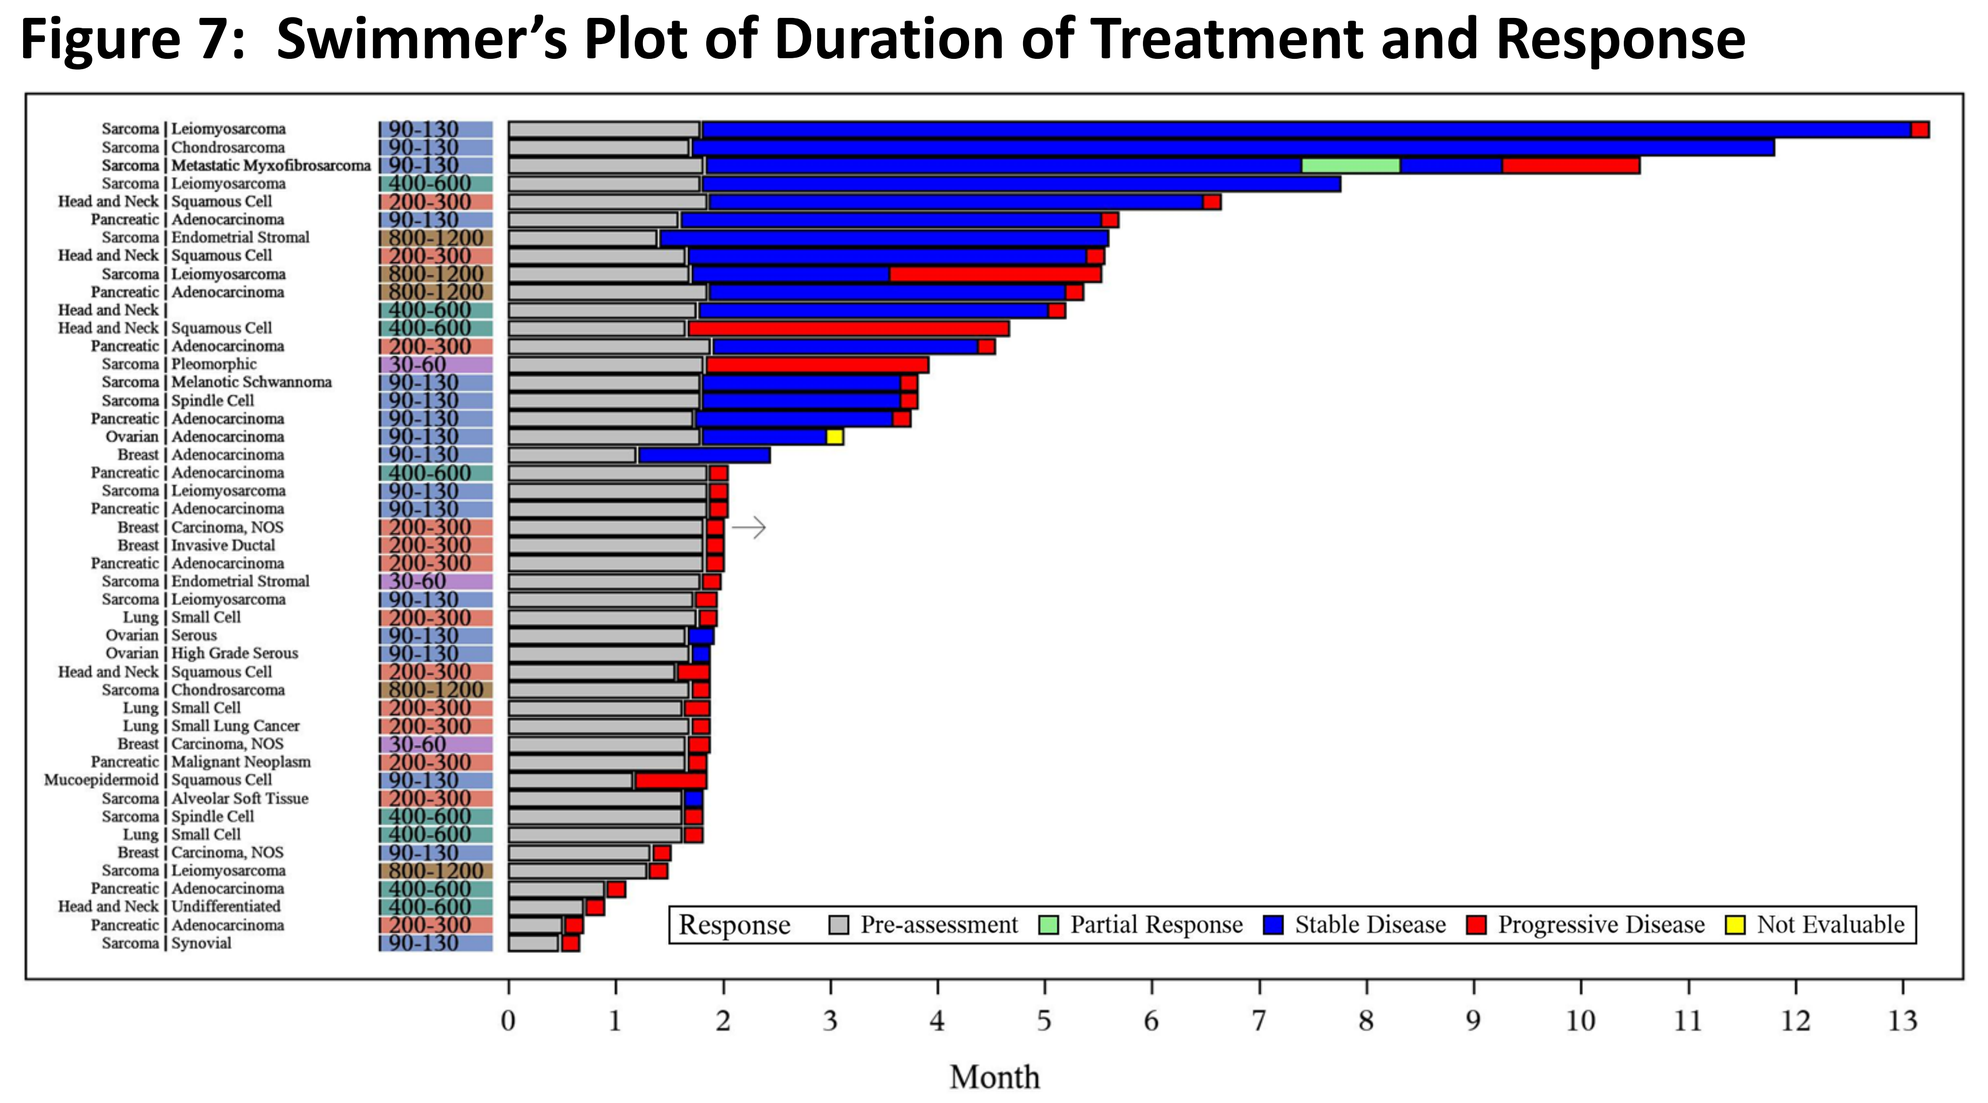
Response
Pre-assessment
Partial Response
Stable Disease
Progressive Disease
Not Evaluable
Month

PR in Sarcoma

Baseline	Month 7

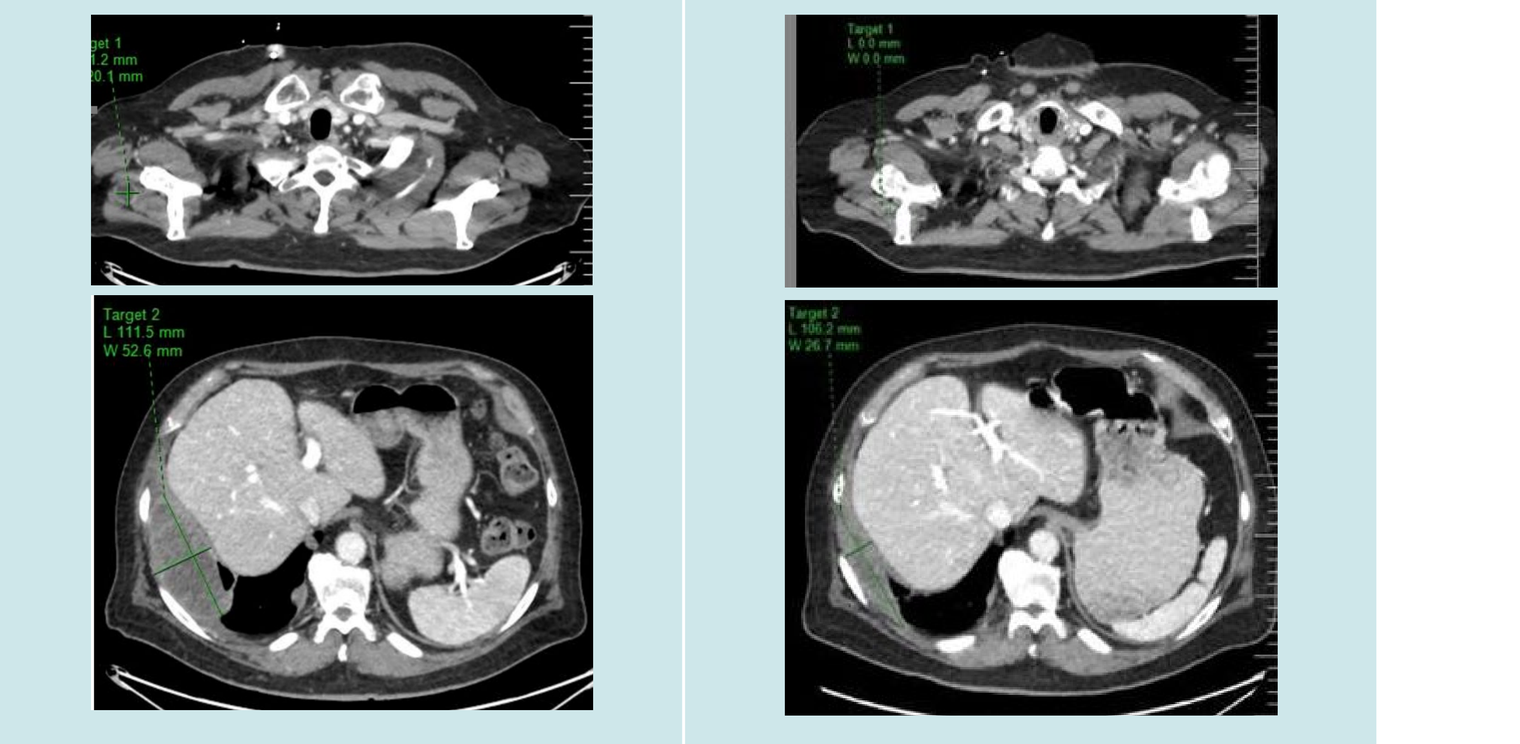

Pt 06: 73-year-old male with myxofibrosarcoma previously treated with 4 lines of therapy. His most recent two therapies were investigational agents with no better response than PD. He was treated with CYT-0851 starting at 45 mg BID and experienced an unconfirmed PR after 7 months. He continued treatment for 11 months until he experienced disease progression due to a new enlarging lymph node. The response was unconfirmed.

Clinical Activity in Lymphoma

Figure 8: Waterfall Plot of Best Percentage Change in Tumor Size

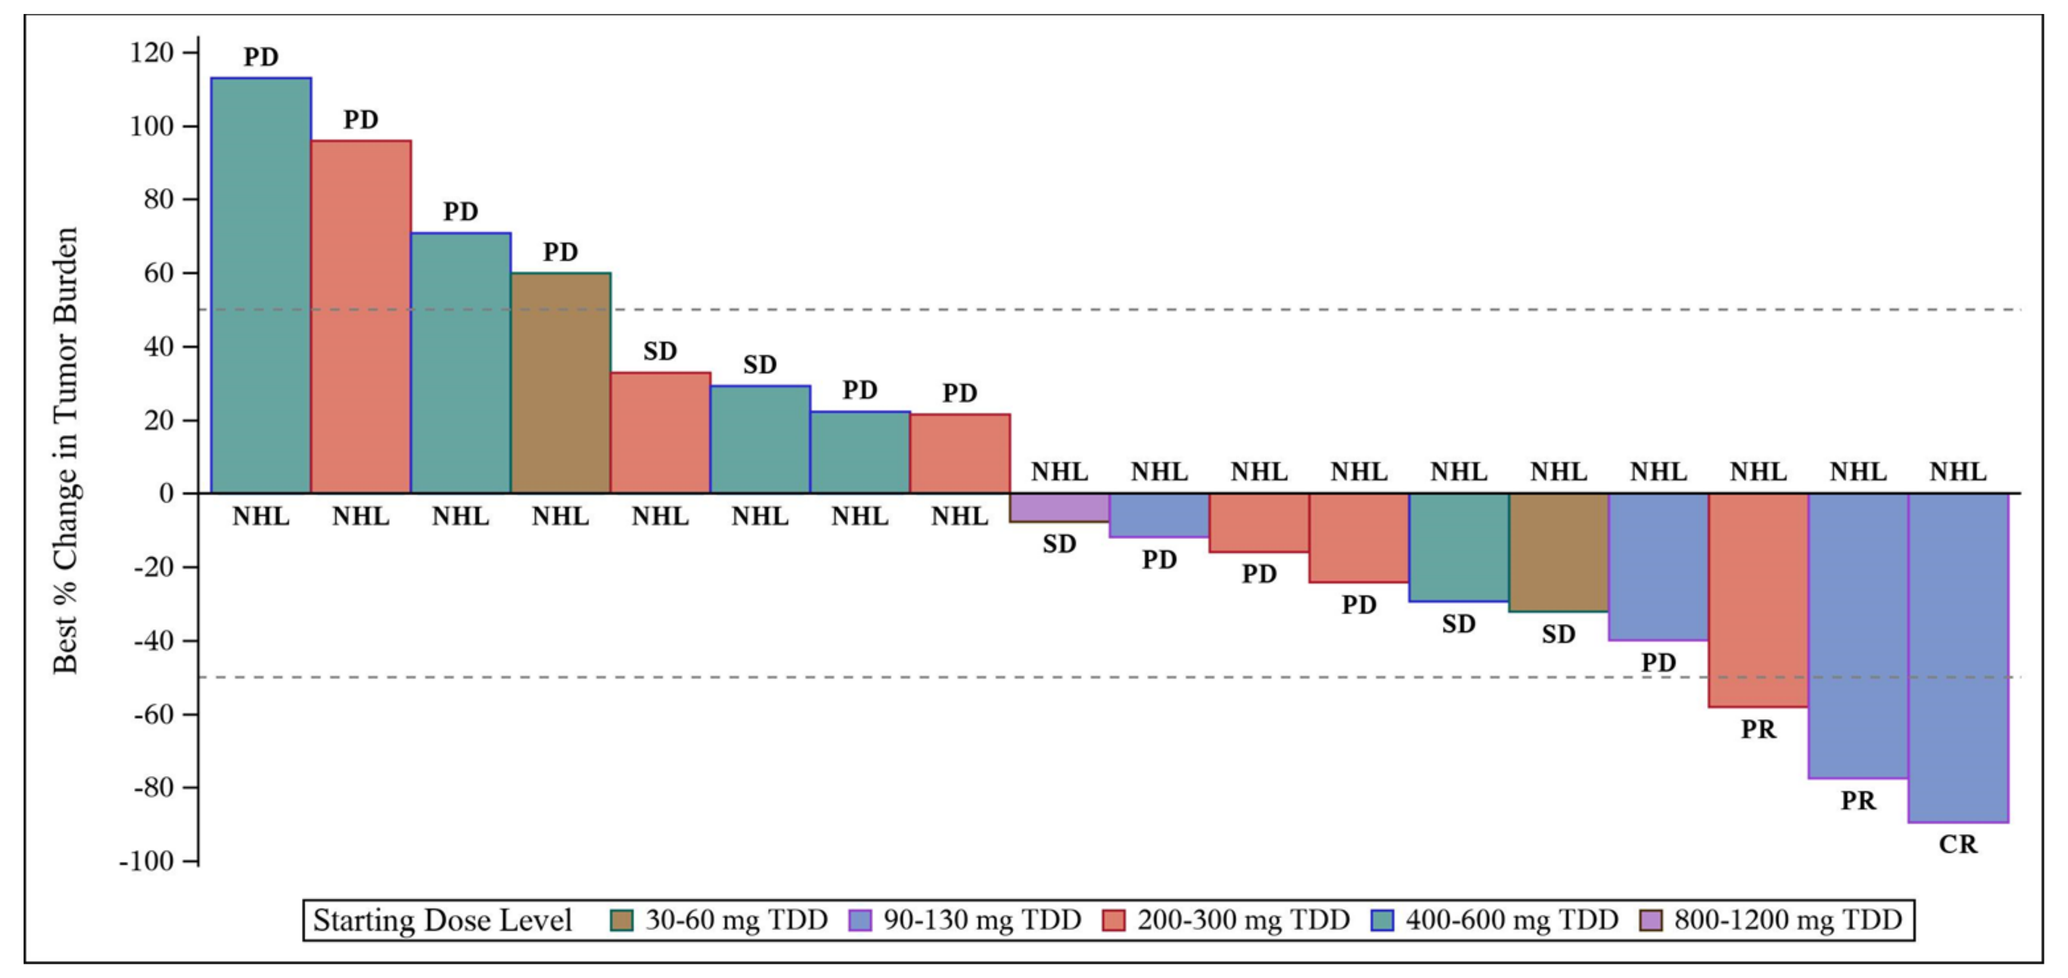
Best % Change in Tumor Burden
Starting Dose Level
30-60 mg TDD
90-130 mg TDD
200-300 mg TDD
400-600 mg TDD
800-1200 mg TDD

Figure 9: Swimmer's Plot of Duration of Treatment and Response

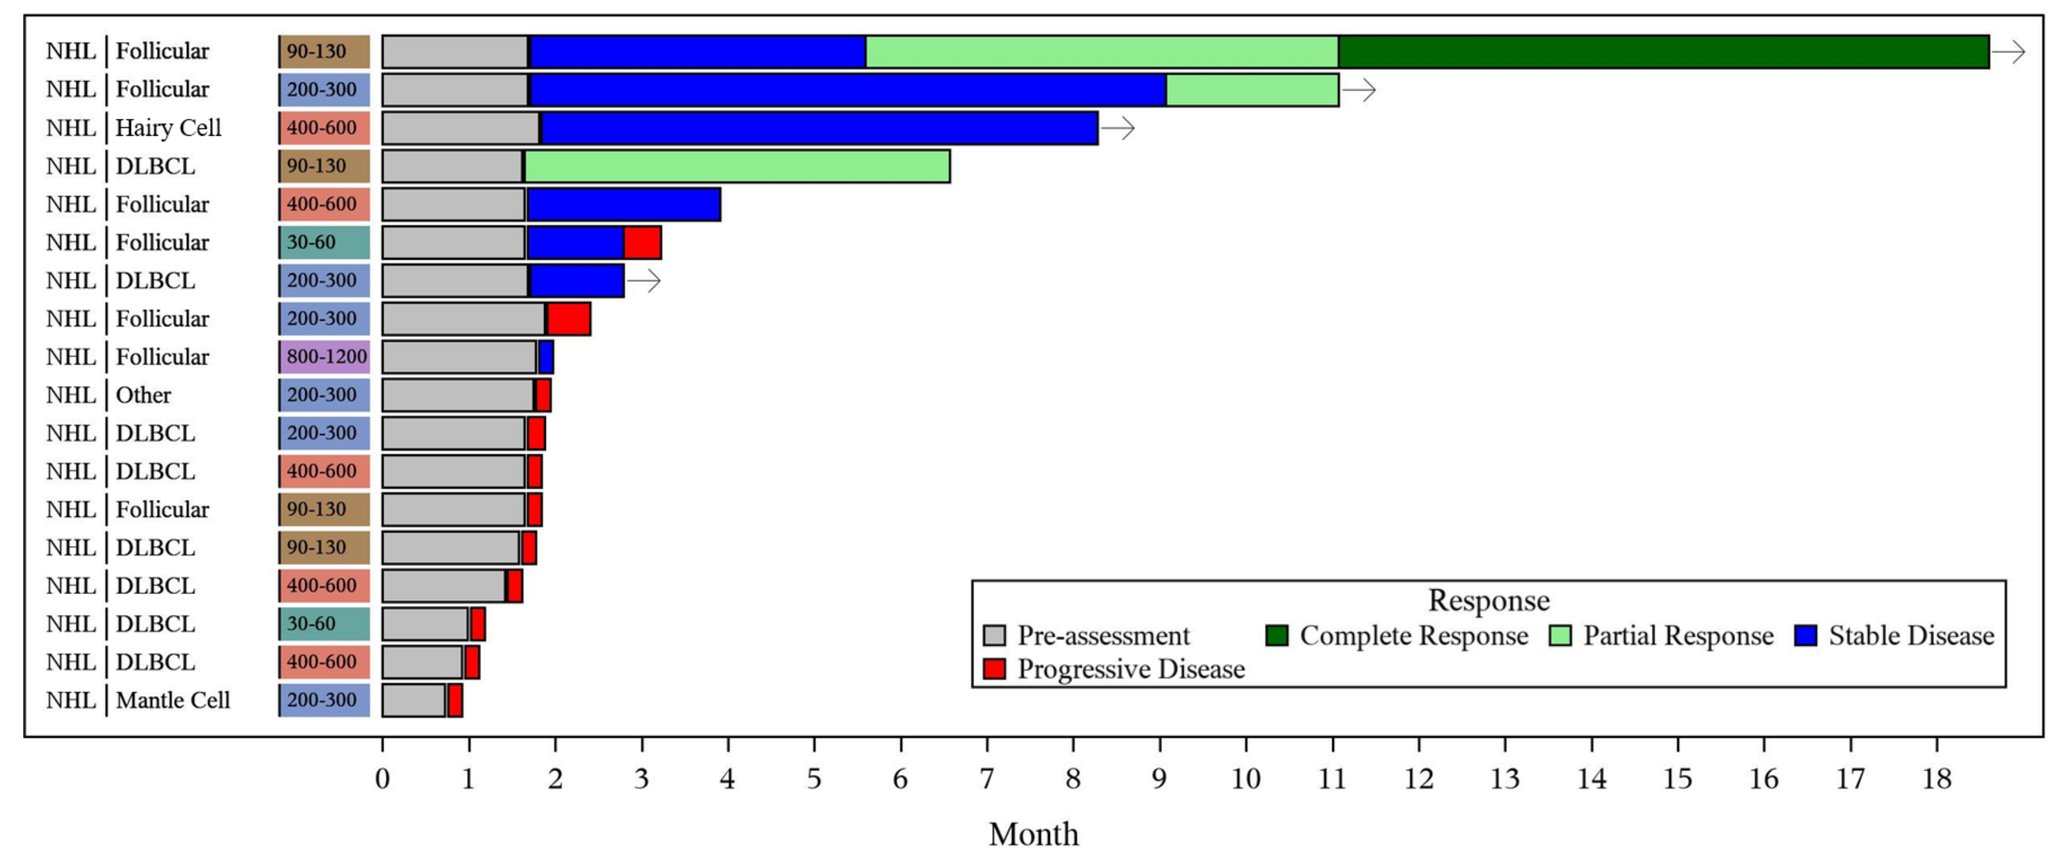
Response
Pre-assessment
Complete Response
Partial Response
Stable Disease
Progressive Disease
Month

- 18 pts with Non-Hodgkin Lymphoma were response evaluable
- Follicular (7 pts): 1 CR at 18+ months, 1 PR at 11+ months; 3 SD; 2 PD
- DLBCL (8 pts): 1 PR;
- 1 SD; 6 PD
- Hairy Cell Leukemia (1 pt): 1 SD ongoing at 8 months, alleviated patients transfusion dependency
- Pts that responded to CYT-0851 experienced durable clinical benefit

CR in Follicular Lymphoma

Baseline	Month 12

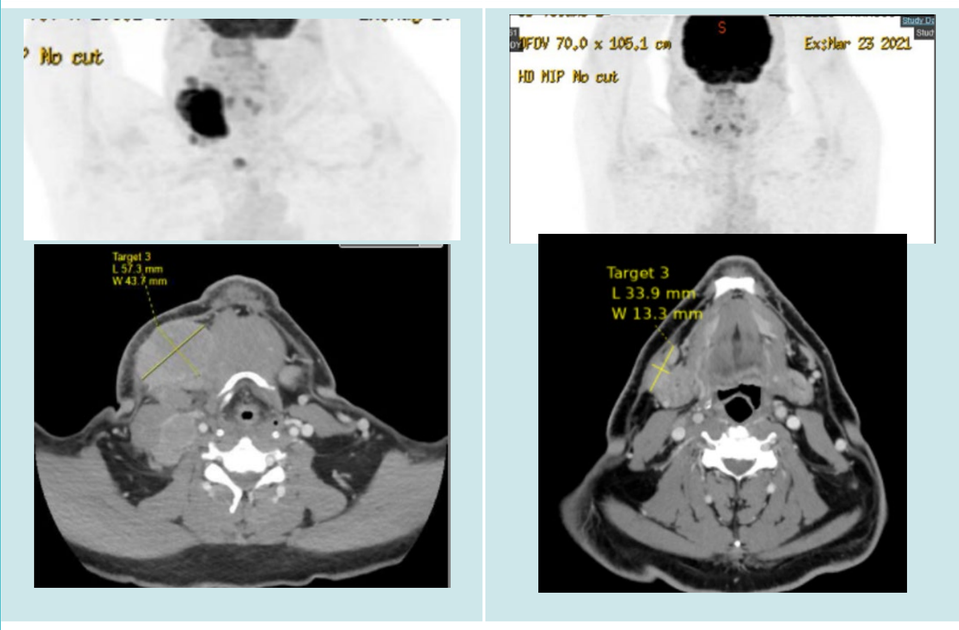

Pt 21: 59-year-old male with follicular lymphoma previously treated with 5 prior lines of therapy. Most recently treated with parsaclisib for 11 months with a best response of PR. He started CYT-0851 at 45 mg BID and experienced a PR after 6 mos and CR after 12 mos on treatment. His treatment is ongoing in Cycle 21.

PR in Follicular Lymphoma

Baseline	Month 11

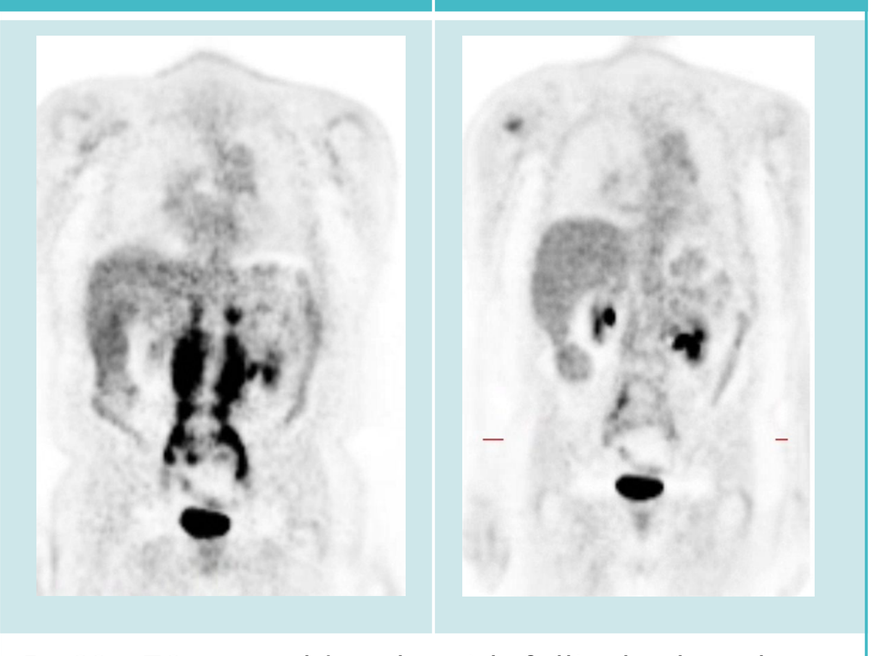

Pt 60: 78-year-old male with follicular lymphoma previously treated with 4 prior lines of therapy. Most recently treated with obinutuzumab (Anti-CD-20) with a best response of PD. Prior to that received Revlimid + rituximab for 12 months with a best response of SD. He started CYT-0851 at 300 mg QD and experienced a PR after 9 months with treatment ongoing in Cycle 11.

PR in DLBCL

Baseline	Month 3

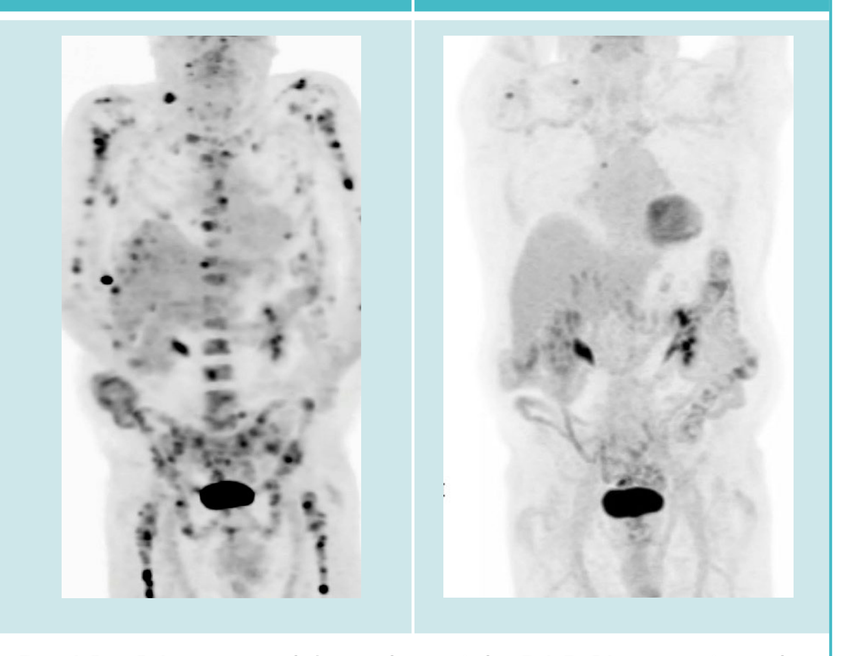

Pt 13: 81-year-old male with DLBCL previously treated with 2 lines of therapy. Most recently treated with rituximab + bendamustine with a PR before quickly progressing. He started CYT-0851 at 45 mg BID and experienced a PR after 2 months. He remained on treatment with systemic control over his disease until month 7 when he developed CNS progression.

Imaging courtesy of Dr. Joel Berman, M.D, Florida Cancer Specialists

Conclusions

- CYT-0851 has demonstrated promising and broad clinical activity in an unselected heavily pre-treated Ph 1 population of patients with hematologic and solid cancers
- CYT-0851 has a generally favorable safety profile characterized by events that were infrequent, primarily Gr1/2, and reversible
- CYT-0851 PK supports QD administration and achieves steady-state levels at doses of 90 mg QD and above that are consistent with tumor growth inhibition in preclinical models
- The MTD for CYT-0851 was determined to be 600 mg QD and a RP2D of 400 mg QD was selected based on the totality of safety and PK data
- The safety and clinical activity observed in the first-in-human trial of CYT-0851 support ongoing evaluation as a monotherapy in 6 expansion cohorts (DLBCL, Follicular Lymphoma, Myeloma, Pancreatic, Ovarian and Soft-Tissue Sarcoma) and in Ph1 combination with 1) gemcitabine, 2) capecitabine and 3) rituximab + bendamustine

Acknowledgments

Cyteir and the investigators want to thank the patients, their families and caregivers for their participation in this study

Presented at the American Society of Clinical Oncology (ASCO) 2022 Annual Meeting, June 3-7, 2022. Contact Information: Dr. Ryan Lynch (rclynch@uw.edu)