363

CYT-0851, a Novel RAD51 Inhibitor, Acts Synergistically with PARP Inhibitors

Tyler Maclay, Melinda Day, and Kevin Mills.
Cyteir Therapeutics, Lexington, MA

Background:

Genomic instability is recognized as a driver of tumorigenesis and cancer progression. Loss of tumor suppressors or activation of oncogenes can induce DNA damage stress, promoting genomic instability and creating dependencies upon key DNA repair pathways. These dependencies can be targeted therapeutically to induce synthetic lethality. We have developed a novel RAD51 inhibitor, CYT-0851, which we have previously shown to be selectively active in Activation Induced Cytidine Deaminase (AID) expressing cells. In cancer cells, AID causes significant genotoxic stress through DNA replication fork collapse which creates a dependency upon the homologous recombination repair factor, RAD51, for survival. CYT-0851 acts by destabilizing RAD51 focus formation, leading to it premature nuclear export and subsequent degradation. PARP inhibitors use another synthetic lethal mechanism, in which PARP1, a protein important for repairing single strand breaks, is inhibited in BRCA1/2 deficient cancers. A main resistance mechanism to PARP inhibitors is the overexpression of RAD51. We therefore hypothesized that our RAD51 inhibitor could act as a sensitizer to PARP inhibitors

Methods:

To determine potential drug synergy, a matrix study was performed with CYT-0851 and 5 different PARP inhibitors including olaparib, niraparib, veliparib, rucaparib, and talazoparib. The combination matrix was tested in 3 cell lines of varying AID expression: ARPE19/HPV16, KYSE-70 and Daudi. PARP indicated cells lines were also tested including HCC1937, HCC1143, and BT20, all of which are derived from triple negative breast cancers and were selected for their varying responsiveness to olaparib or BRCA status.

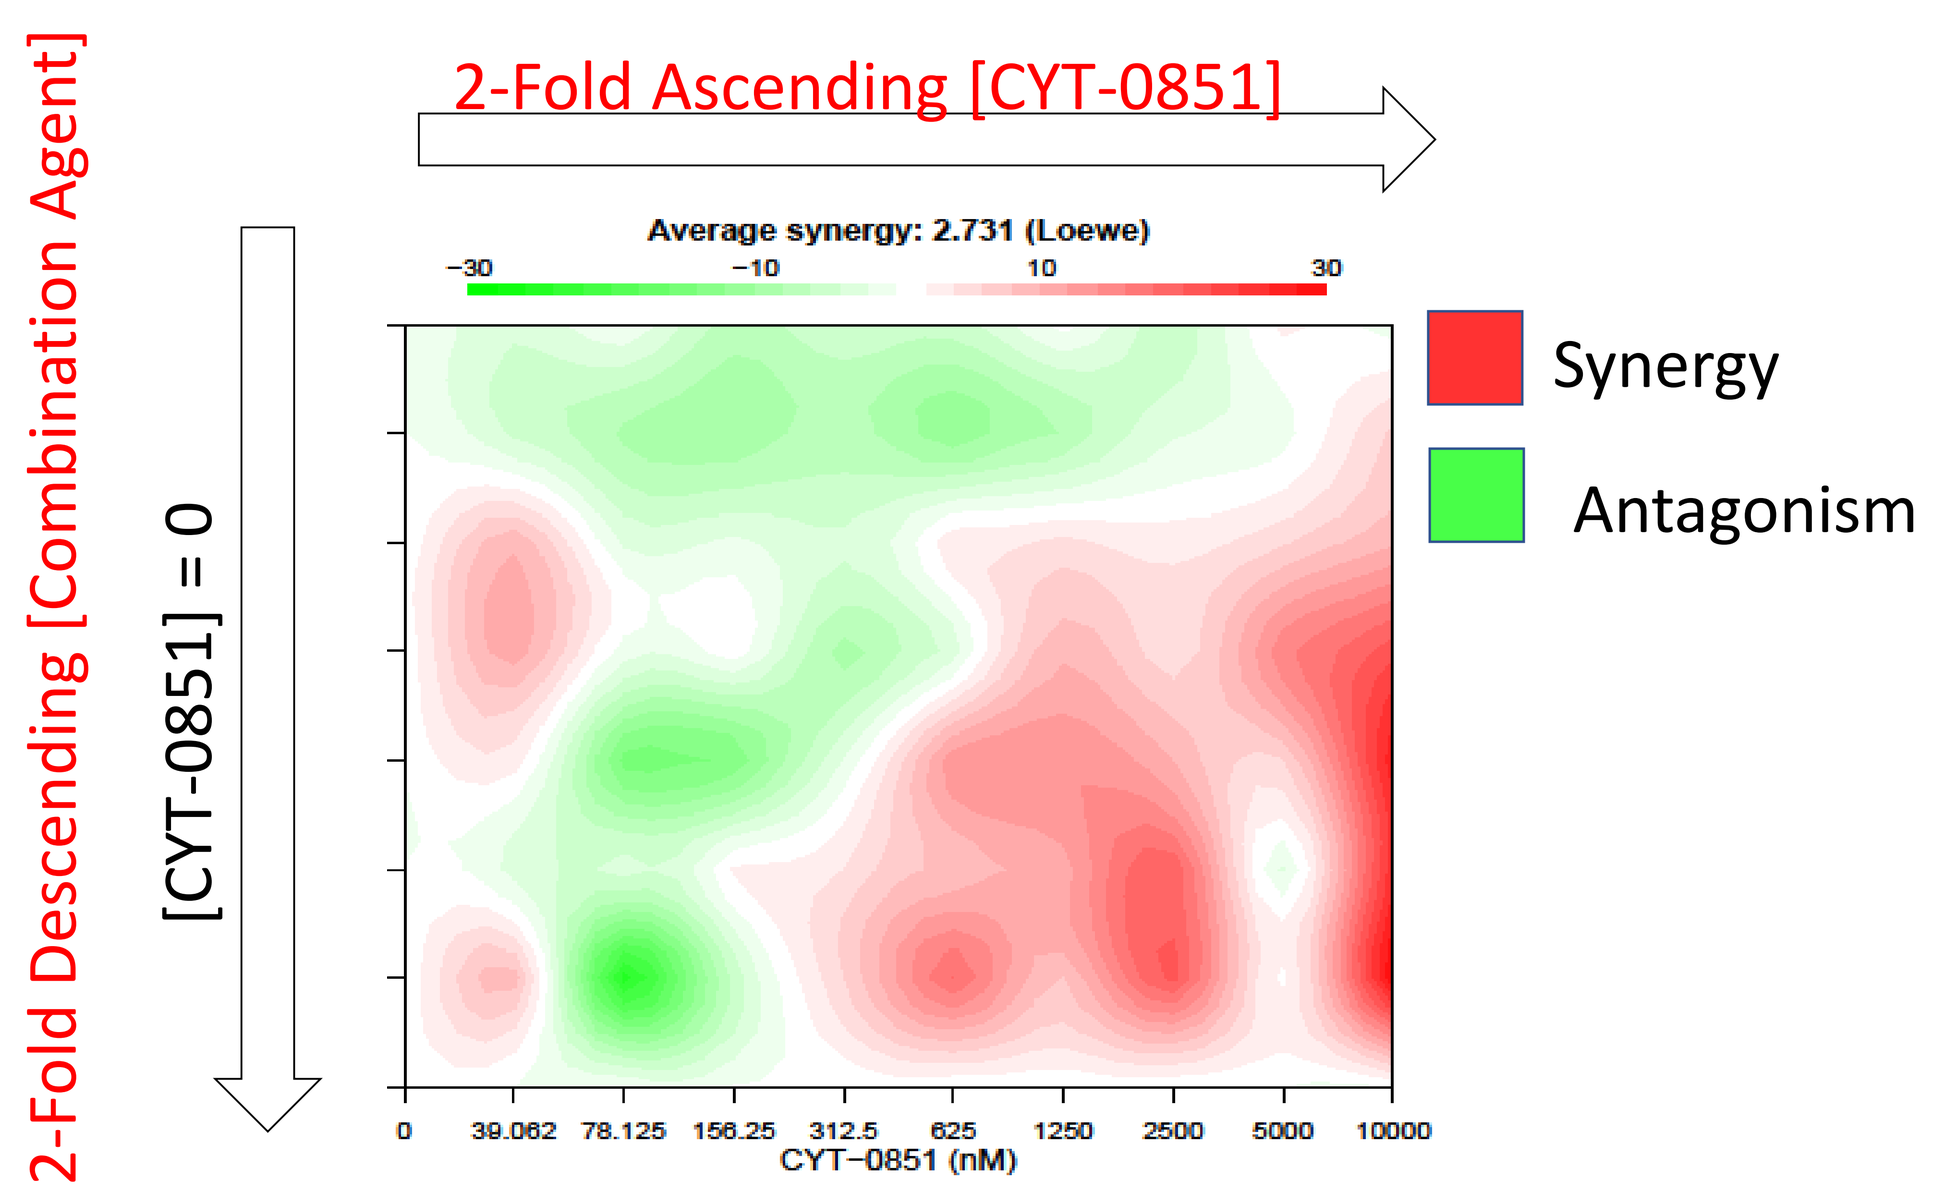
2-Fold Ascending [CYT-0851]
2-Fold Descending [Combination Agent]
[CYT-0851] = 0
Average synergy: 2.731 (Loewe)
CYT−0851 (nM)
[Combination Agent] = 0
Synergy
Antagonism

Drug	[High], uM	[Low], uM
CYT-0851	5	.020
Olaparib	5	.078
Niraparib	5	.078
Veliparib	50	.781
Rucaparib	10	.156
Talazoparib	.625	.009

Models:

Bliss Independence model was used and calculated in Excel. The model assumes that the drugs act with probabilistic independence.

$$\text{Bliss: } y_{\text{exp}} = y_A + y_B - y_A y_B$$

This expected value was generated by examining single-agent effects and then tested against the observed value using a two-tailed Welch's T-Test. For visualization purposes, the values generated from this test were transformed to a "SynergyScore" by doing the computation:

$$\text{SynergyScore} = \frac{-\log(p)}{\log(0.05)} x \frac{t}{|t|}$$

Loewe Additivity model was used and calculated by the "Synergyfinder" R package.

$$\frac{x_1}{f_1^{-1}(y_{Loewe})} + \frac{x_2}{f_2^{-1}(y_{Loewe})} = 1$$

The graphs generated describe the difference between the expected value generated by the additivity model and the observed value generated by the experiment. If the effect determined by the additivity model < the effect observed the space is colored red, if the effect determined by the additivity mode > the effect observed then the space is colored green. The saturation of the color is proportional to the magnitude of the difference between these two values.

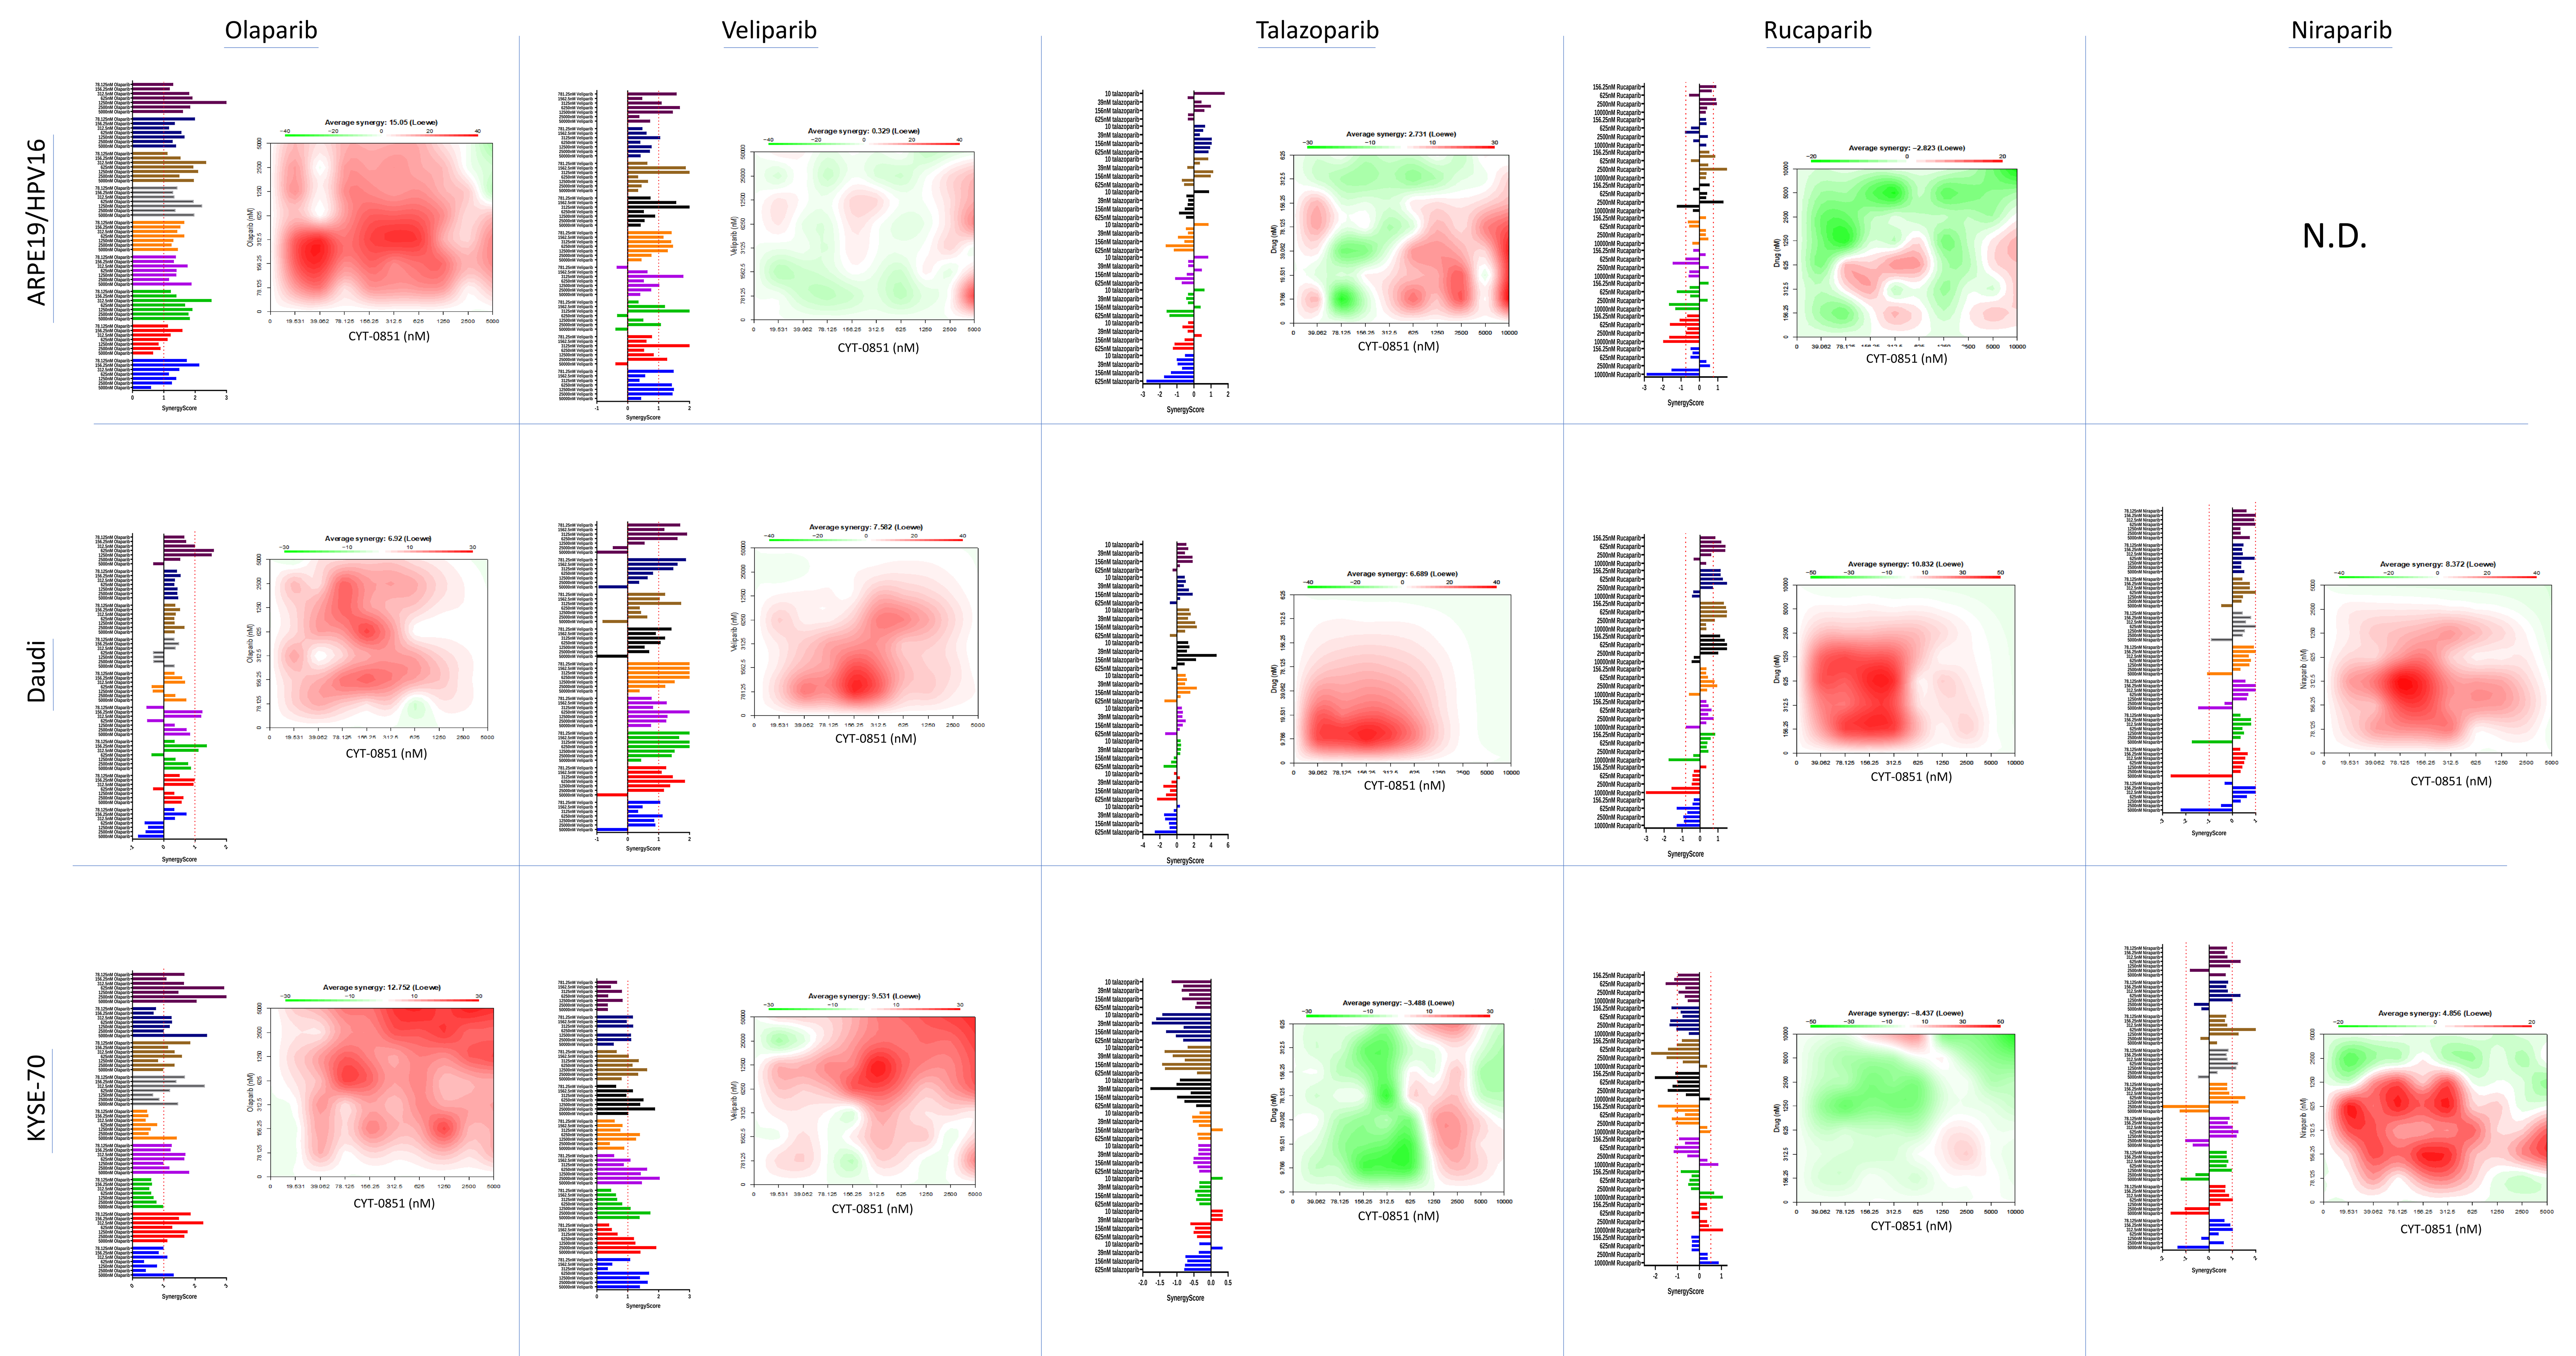
Olaparib
Veliparib
Talazoparib
Rucaparib
Niraparib
ARPE19/HPV16
Daudi
KYSE-70
N.D.
Average synergy: 15.05 (Loewe)
Average synergy: 0.329 (Loewe)
Average synergy: 2.731 (Loewe)
Average synergy: −2.823 (Loewe)
Average synergy: 6.92 (Loewe)
Average synergy: 7.582 (Loewe)
Average synergy: 6.689 (Loewe)
Average synergy: 10.832 (Loewe)
Average synergy: 8.372 (Loewe)
Average synergy: 12.752 (Loewe)
Average synergy: 9.531 (Loewe)
Average synergy: −3.488 (Loewe)
Average synergy: −8.437 (Loewe)
Average synergy: 4.856 (Loewe)
CYT-0851 (nM)
SynergyScore

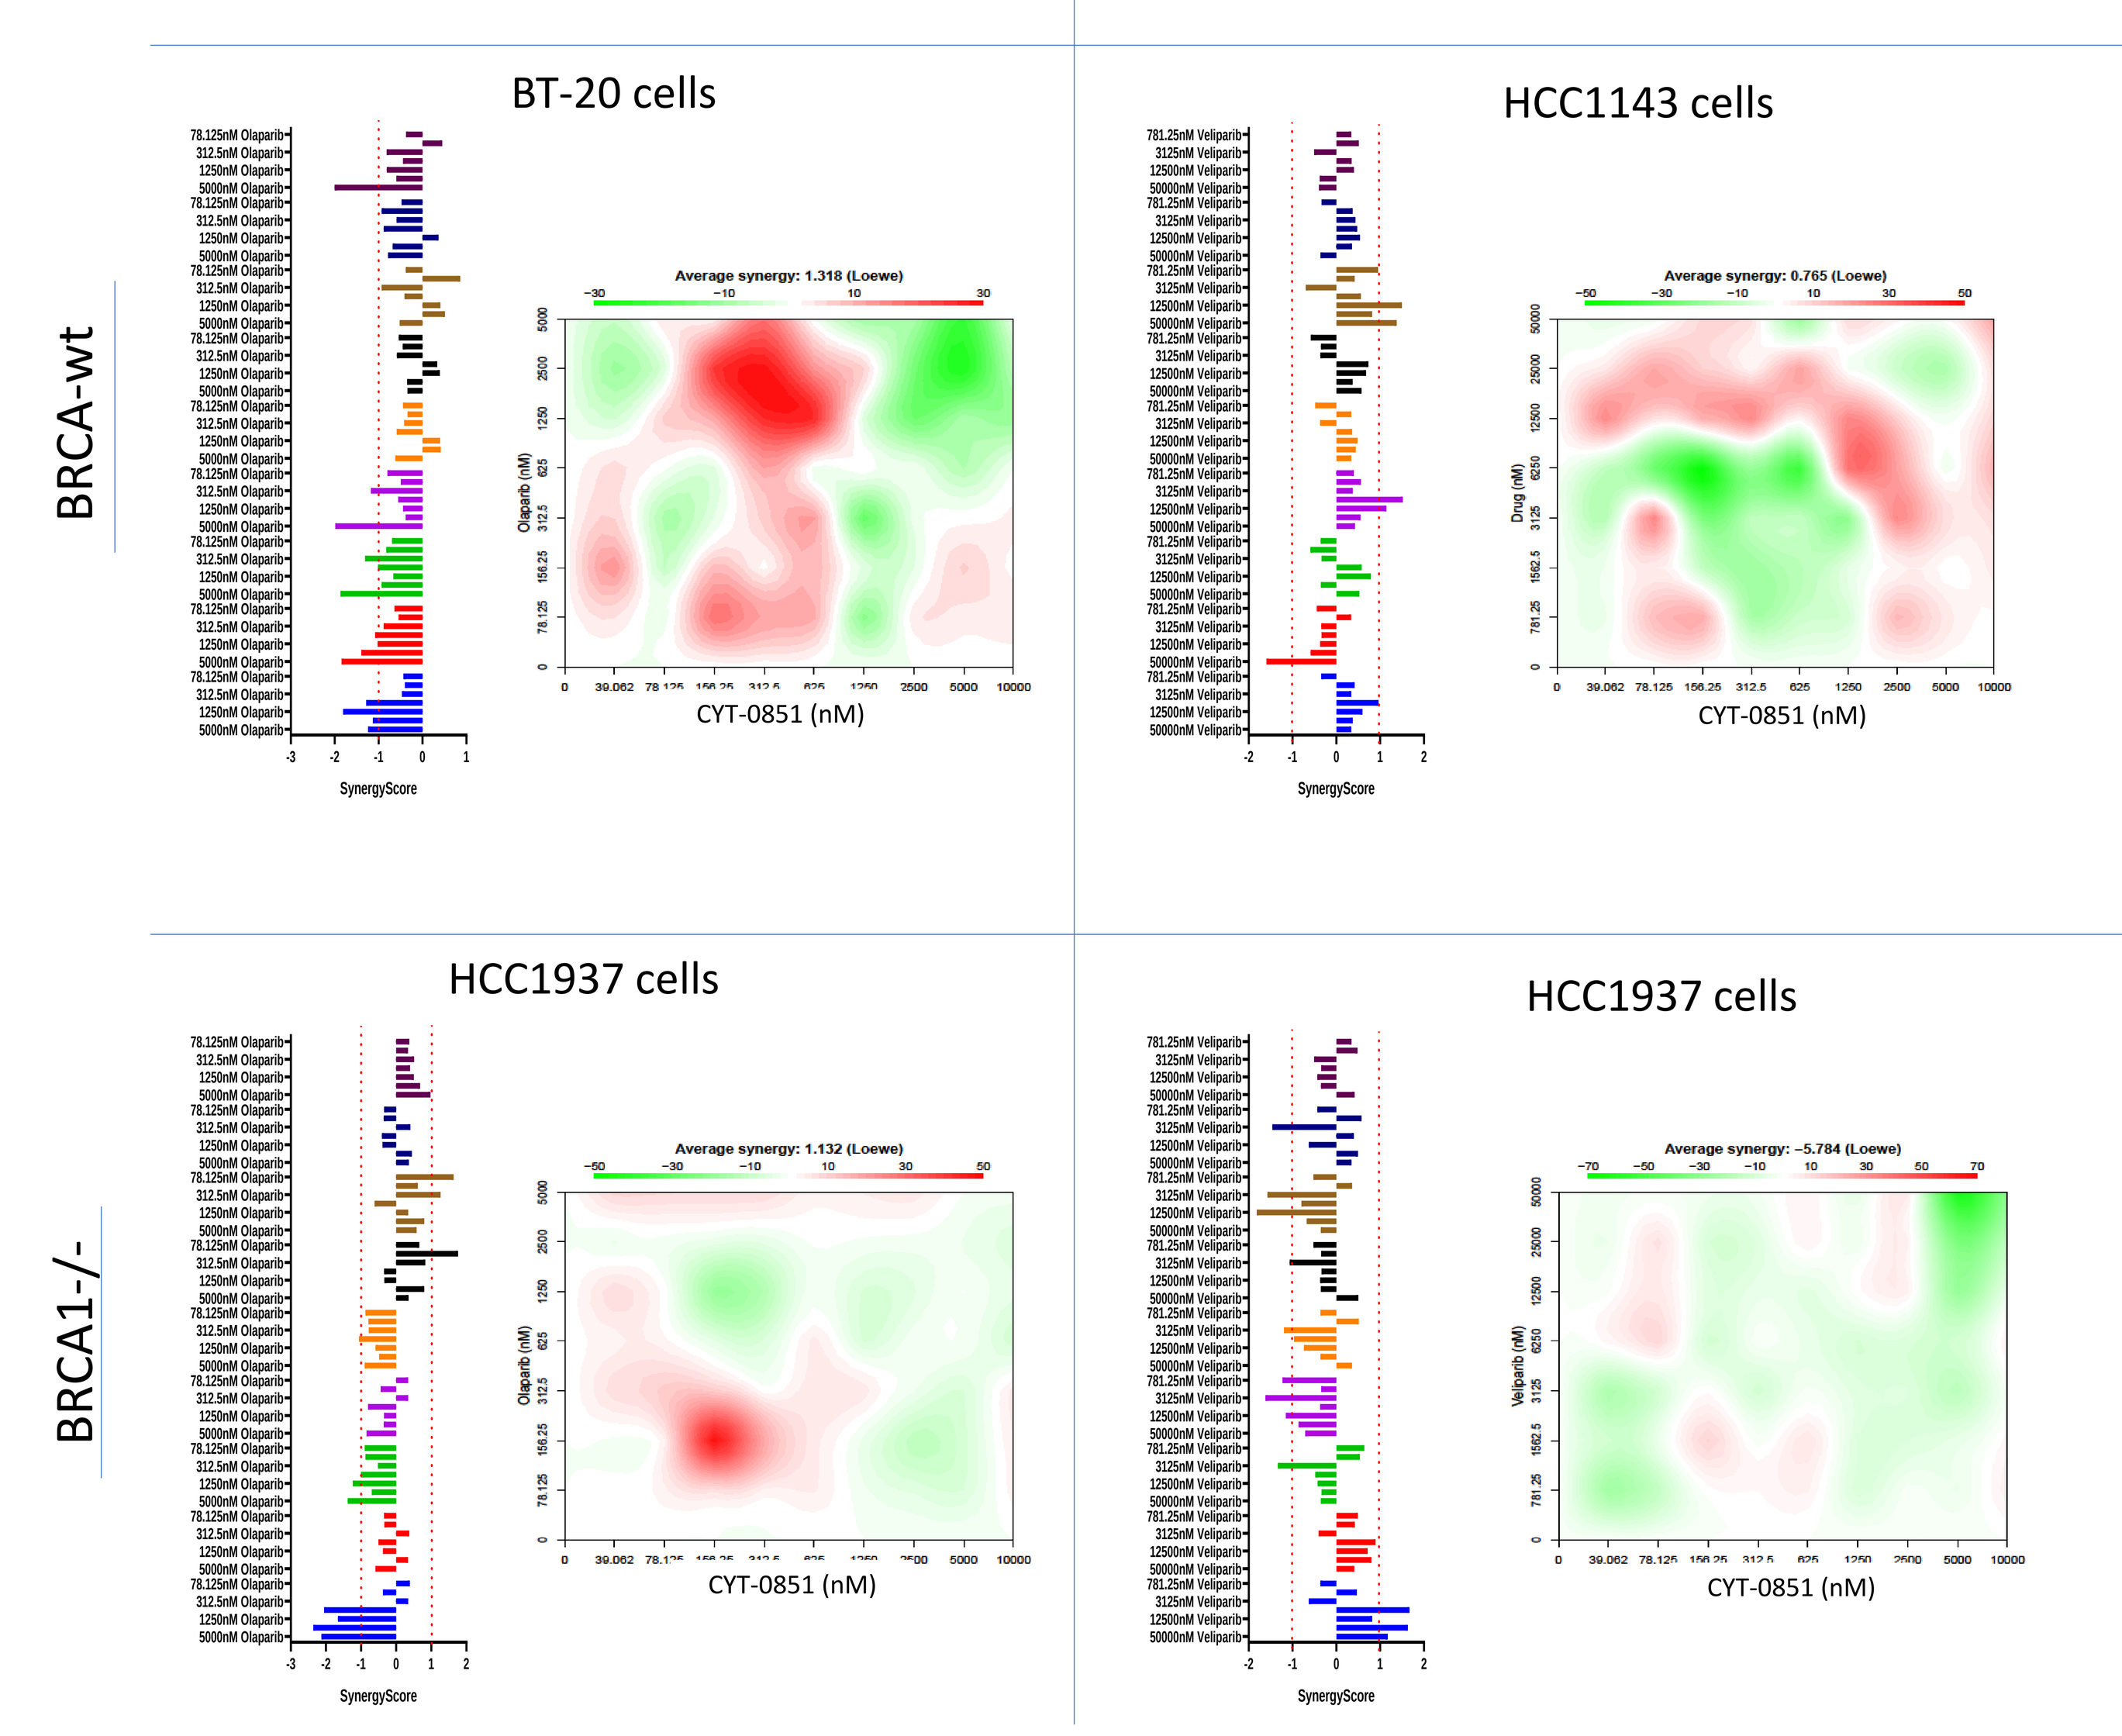
BRCA-wt
BRCA1-/-
BT-20 cells
HCC1143 cells
HCC1937 cells
HCC1937 cells
Average synergy: 1.318 (Loewe)
Average synergy: 0.765 (Loewe)
Average synergy: 1.132 (Loewe)
Average synergy: −5.784 (Loewe)
CYT-0851 (nM)
SynergyScore

Barchart Key

- 5000nM CYT-0851
- 2500nM CYT-0851
- 1250nM CYT-0851
- 625nM CYT-0851
- 312nM CYT-0851
- 156nM CYT-0851
- 78nM CYT-0851
- 39nM CYT-0851
- 19nM CYT-0851

Cell Line	Cell Type
ARPE19/HPV16	AID negative
Daudi	AID-High
KYSE-70	AID-Low
HCC1143	Breast
HCC1937	Breast (BRCA1-/-)
BT-20	Breast

Figures:

- Heat maps represent Loewe additivity model calculations. Red colored regions are synergistic, green colored regions are antagonistic, and white/unsaturated regions are additive.
- Bar charts show Bliss Independence model calculations, the dotted red bar shows the region where P < 0.05 by 2-tailed Welch's t-test.

Summary of Results:

Drug/Combination	Cell Conditions Tested	Conclusion
Olaparib	AID expression range (Low, Med, High), BRCA-Status (wt, -/-)	All cells tested showed strong synergy with olaparib independent of AID expression. In both BRCA-wt/BRCA-mut conditions combination treatment showed synergy.
Veliparib	AID expression range (Low, Med, High), BRCA-Status (wt, -/-)	All cells tested showed strong synergy with veliparib independent of AID expression. In both BRCA-wt/BRCA-mut conditions combination treatment showed synergy.
Rucaparib	AID expression range (Low, Med, High)	The combination was synergistic only in the AID-high cells at low concentrations.
Talazoparib	AID expression range (Low, Med, High)	The combination was synergistic in all AID positive conditions.
Niraparib	AID expression range (Low, Med, High)	The combination was strongly synergistic in all AID positive conditions.

- **CYT-0851 showed synergy with all PARP inhibitors tested**
- **Degree of synergy between CYT-0851 and $PARP_i$ was correlated with the reported PARP trapping activity**
- **Synergy was observed in BRCA-wt breast cancer lines, in which $PARP_i$ is not effective as a single agent**

References:

- Ianevski A, He L, Aittokallio T, Tang J. SynergyFinder: a web application for analyzing drug combination dose-response matrix data. Bioinformatics. 2017;33(15):2413-2415.
- Tang J, Wennerberg K, Aittokallio T. What is synergy? The Saariselkä agreement revisited. Front Pharmacol. 2015;6:181. Published 2015 Sep 1. doi:10.3389/fphar.2015.00181

*TM, MD, and KM are employees at Cyteir Therapeutics, JV, CM, and AC are paid consultants at Cyteir Therapeutics

AACR 2019